EFFECTIVENESS AND SAFETY OF A DERMOCOSMETIC CREAM CONTAINING SALYCILIC ACID, LIPOHYDROXY ACID, NIACINAMIDE, AQUA-POSAE-FILIFORMIS, PROCERAD AND ZINC-PCA AS AN ADJUVANT TREATMENT FOR MILD AND MODERATE ACNE IN INDONESIA

Irma Bernadette S. Sitohang[1,2], Lilik Norawati[3], Satya Wydya Yenny[4], Arie Kusumawardani[5], Sinta Murlistyarini[6], Silvia Veronica Setiawan[3], Aria Kekalih[7]

[1]Division of Cosmetic Dermatology, Department of Dermatology and Venereology, Faculty of Medicine, Universitas Indonesia, Dr. Cipto Mangunkusumo Hospital - Jakarta, Indonesia. [2]Medical Staff Unit of Dermatology and Venereology, Faculty of Medicine, Universitas Indonesia, Universitas Indonesia Hospital - Depok, Indonesia. [3]Department of Dermatology and Venereology, Indonesia Presidential Hospital Gatot Soebroto - Jakarta, Indonesia. [4]Division of Cosmetic Dermatology, Department of Dermatology and Venereology, Faculty of Medicine, Universitas Andalas, Dr. M Djamil Central General Hospital - Padang, Indonesia. [5]Division of Cosmetic Dermatology, Department of Dermatology and Venereology, Faculty of Medicine, Universitas National Surakarta, Dr. Moewardi Regional General Hospital – Surakarta, Indonesia. [6]Division of Cosmetic Dermatology, Department of Dermatology and Venereology, Faculty of Medicine, Universitas Brawijaya, Dr. Saiful Anwar Regional General Hospital – Malang, Indonesia. [7]Department of Community Medicine, Faculty of Medicine, Universitas Indonesia - Jakarta, Indonesia

INTRODUCTION

Acne is a chronic inflammatory condition in which dermocosmetics can be used as an adjuvant treatment to standard therapy with additive benefits in terms of efficacy or tolerability.

The aim of this study was to evaluate the efficacy and tolerability of a dermocosmetic (DC) cream containing Salicylic Acid, Lipohydroxy Acid, Niacinamide, *Aqua-Posae-Filiformis* (APF), Procerad and Zinc-PCA in adjunct to Adapalene in Indonesian patients with mild to moderate acne.

MATERIAL & METHODS

This multicenter, randomized, evaluator-blind, parallel-group study was conducted in five hospitals in Indonesia from May to December 2022. Subjects with mild and moderate acne, aged 15 to 50 years, were randomized into three groups.

GROUP A:	GROUP B:	GROUP C:
0,1% adapalene cream only	0.1% adapalene cream every other night and DC every morning	0.1% adapalene cream every night and DC every morning

Subjects were evaluated on day 28 and day 56. Evaluations included GEA scale and lesion count (IAEM scale), sebum levels recorded using a A-One Simple Ver. 2.53 one-click automatic full-facial skin analysis (BOMTECH ELECTRONICS Co., Ltd., Korea), patient's quality of life (QoL) using CADI and Acne QoL questionnaires, as well as patient's satisfaction (score 1-5, with 1 the lowest) and tolerability (score 1-4, with 1 the least tolerable) evaluated separately by investigator and patient.

RESULTS

Overall, 293 subjects were included in the study and randomized into three treatment groups. Subjects were recruited from 5 different research centers, (54-67 subjects per center). All the subjects were of Asian ethnicity, the majority of them were females (60%), phototypes IV (79.5%), aged >25 y.o. (55.3%), Javanese (54%), with higher education (54%). After 56 days, all three groups showed significant improvements of GEA scale, lesion count and QoL. Group C showed a higher reduction of GEA score compared to Group A (ANOVA repeated measures ; p=0.038). No significant difference was found on sebum levels among the three groups. Patient's satisfaction and tolerability were evaluated separately by both investigators and subjects. There were significant differences in investigator's evaluation of tolerability among the three groups (p 0.001), with group A (adapalene alone) having higher score (less tolerable) than both groups B and C on day 28 and day 56. For subject's evaluation of tolerability, no difference was observed between groups B and C.
Evaluation of subject's satisfaction by the investigators was significantly different between the three groups with groups B and C having higher scores than group A both on day 28 and day 56. Even if an increase was observed in group A at day 56, no significant difference was observed between groups B and C for the evaluation of satisfaction by the subjects. In group B, there was significant increase in subjects scoring with higher scores between day 28 and day 56.

Figure 1 : Subject from Group C showed visible improvement from day 0 to day 28 to day 56

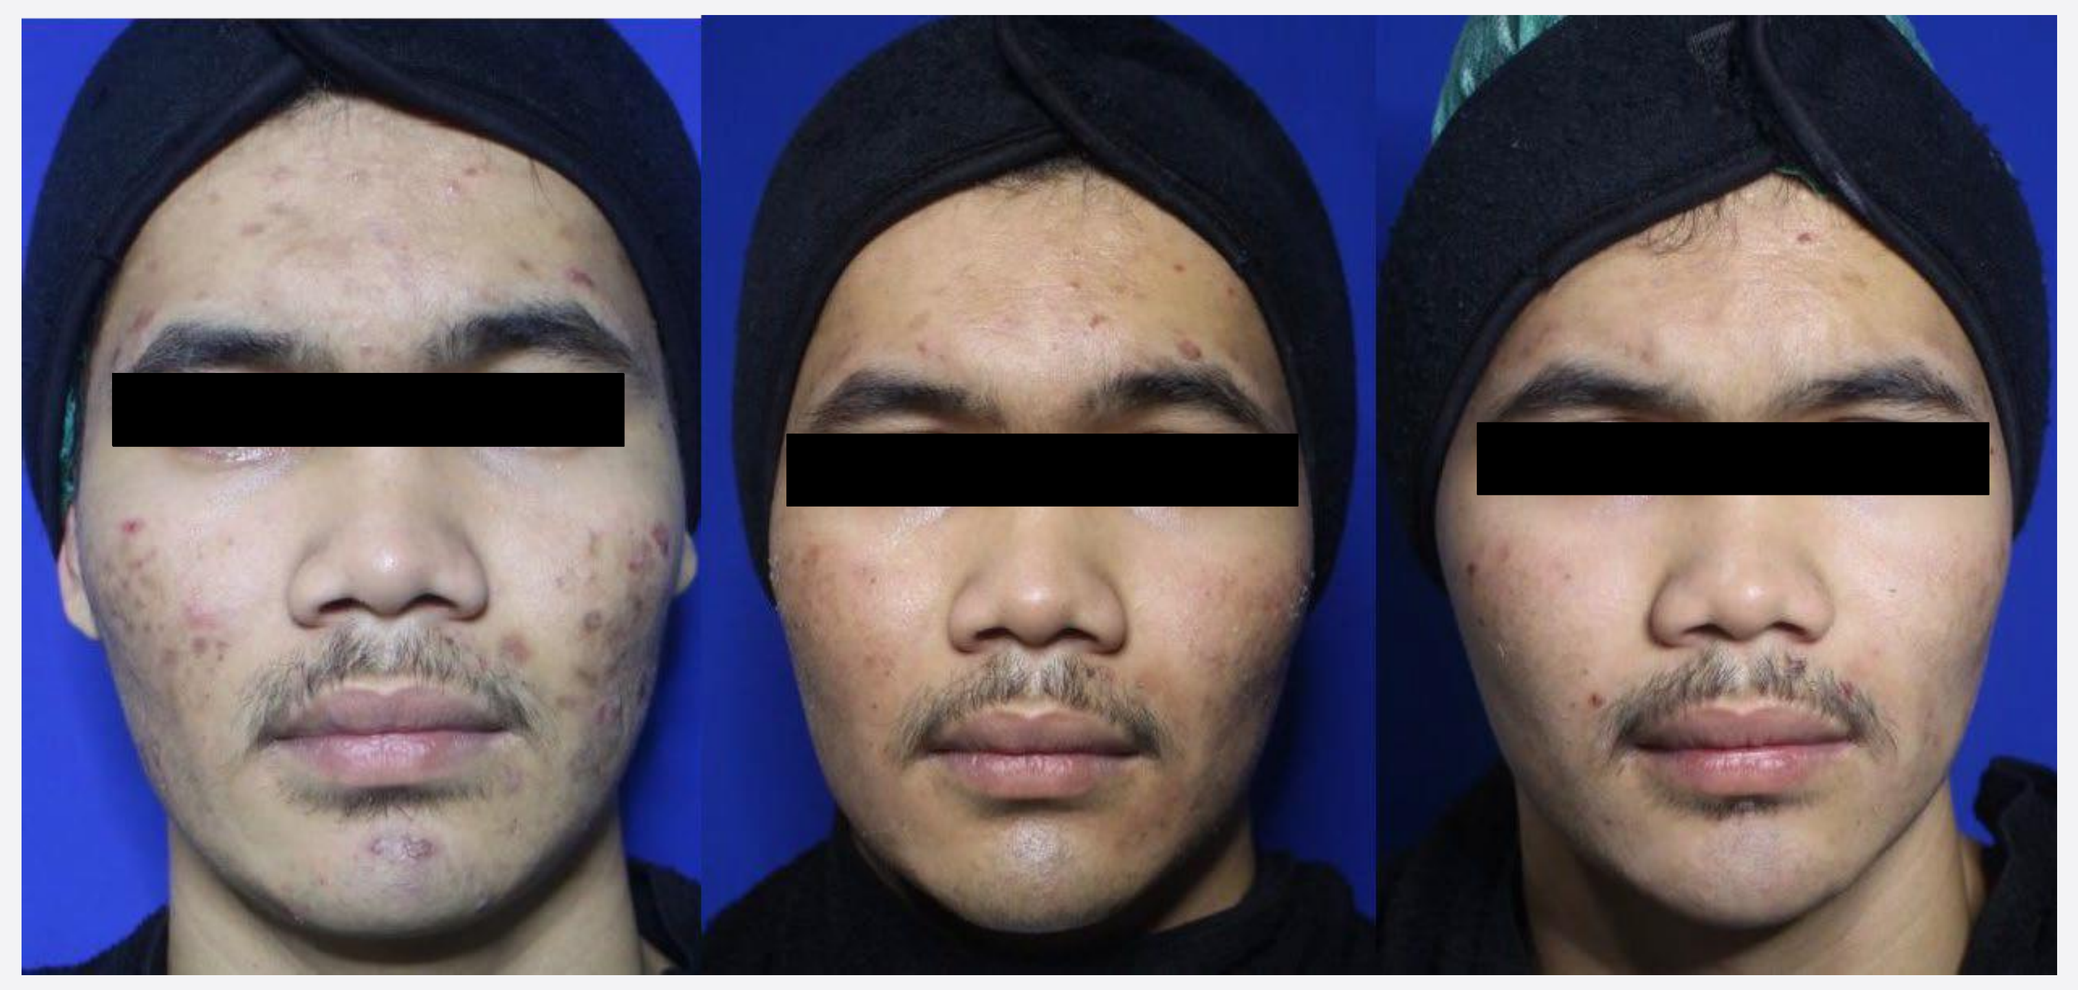

Day 0 Day 28 Day 56

CONCLUSION

The usefulness of an active dermocosmetic as an adjunct to drugs has already been reported in the literature in the management of acne. This is the first time a randomized controlled clinical study is conducted combining a multitargeted dermocosmetic cream with adapalene in mild to moderate acne. This study showed a significant improvement of acne over time in the 3 groups with a superior efficacy of the association of DC and adapalene applied once daily on GEA scale, as well as a superior tolerability and satisfaction for the 2 regimens combining the DC and adapalene compared to adapalene alone.

Acknowledgements:
Dr. Cipto Mangunkusumo Hospital | Indonesia Presidential Hospital Gatot Soebroto | Dr. M Djamil Central General Hospital Dr. Moewardi Regional General Hospital | Dr. Saiful Anwar Regional General Hospital Sponsored by La Roche-Posay – L' Oréal