International dermocosmetic skincare consensus using the RAND/UCLA appropriateness method

Zoe D. DRAELOS MD,[1] Liu WEI MD,[2] Mukta SACHDEV MD,[3] Bruna S. F. BRAVO MD,[4] Vasanop VACHIRAMON MD,[5] Marie JOURDAN MD,[6] Martina KERSCHER MD PhD,[7] Catherine DELVA,[8] Stéphanie LECLERC-MERCIER MD[9]

[1]Dermatology Consulting Services, PLLC, High Point, NC, USA; [2]Department of Dermatology, The General Hospital of Air Force PLA, Beijing, China; [3]Dept of Dermatology, Manipal Hospital, Bangalore, India; MS Clinical Research Pvt Ltd, Bangalore, India; [4]Clinica Bravo and Bravo Research Center, Rio de Janeiro, Brazil; [5]Division of Dermatology, Department of Medicine, Faculty of Medicine Ramathibodi Hospital, Mahidol University, Bangkok, Thailand; [6]Centre Laser International de la Peau-Paris (CLIPP), Paris, France; [7]Divison of Cosmetic Sciences, University of Hamburg, Germany; [8]Inferential, Paris, France; [9]Laboratoires Vichy International, Levallois-Perret, France.

1 INTRODUCTION & OBJECTIVES

- Consumers often seek recommendations from dermatologists on the best ingredients in dermocosmetics.
- Recommendations can be introduced gradually starting with essential products for protection and repair, then renewal, and finally stimulation depending on patients' specific concerns.[1,2]
- The objective of this international expert consensus was to provide a tool for recommendations on anti-aging dermocosmetics for diverse patient populations across all phototypes, both genders and with exposure to various exposome factors.

2 MATERIALS & METHODS

- 7 international experts reviewed 8 hypothetical case scenarios.
- The recommendations were extended to cover both genders and all Fitzpatrick skin types (FST).
- The RAND/UCLA appropriateness method was used to obtain consensus.
- The experts completed a questionnaire to evaluate the appropriateness of 17 key ingredients in dermocosmetics on a scale from 1 (totally inappropriate) to 9 (totally appropriate).
- Two meetings and email discussions refined the recommendations.

3 RESULTS

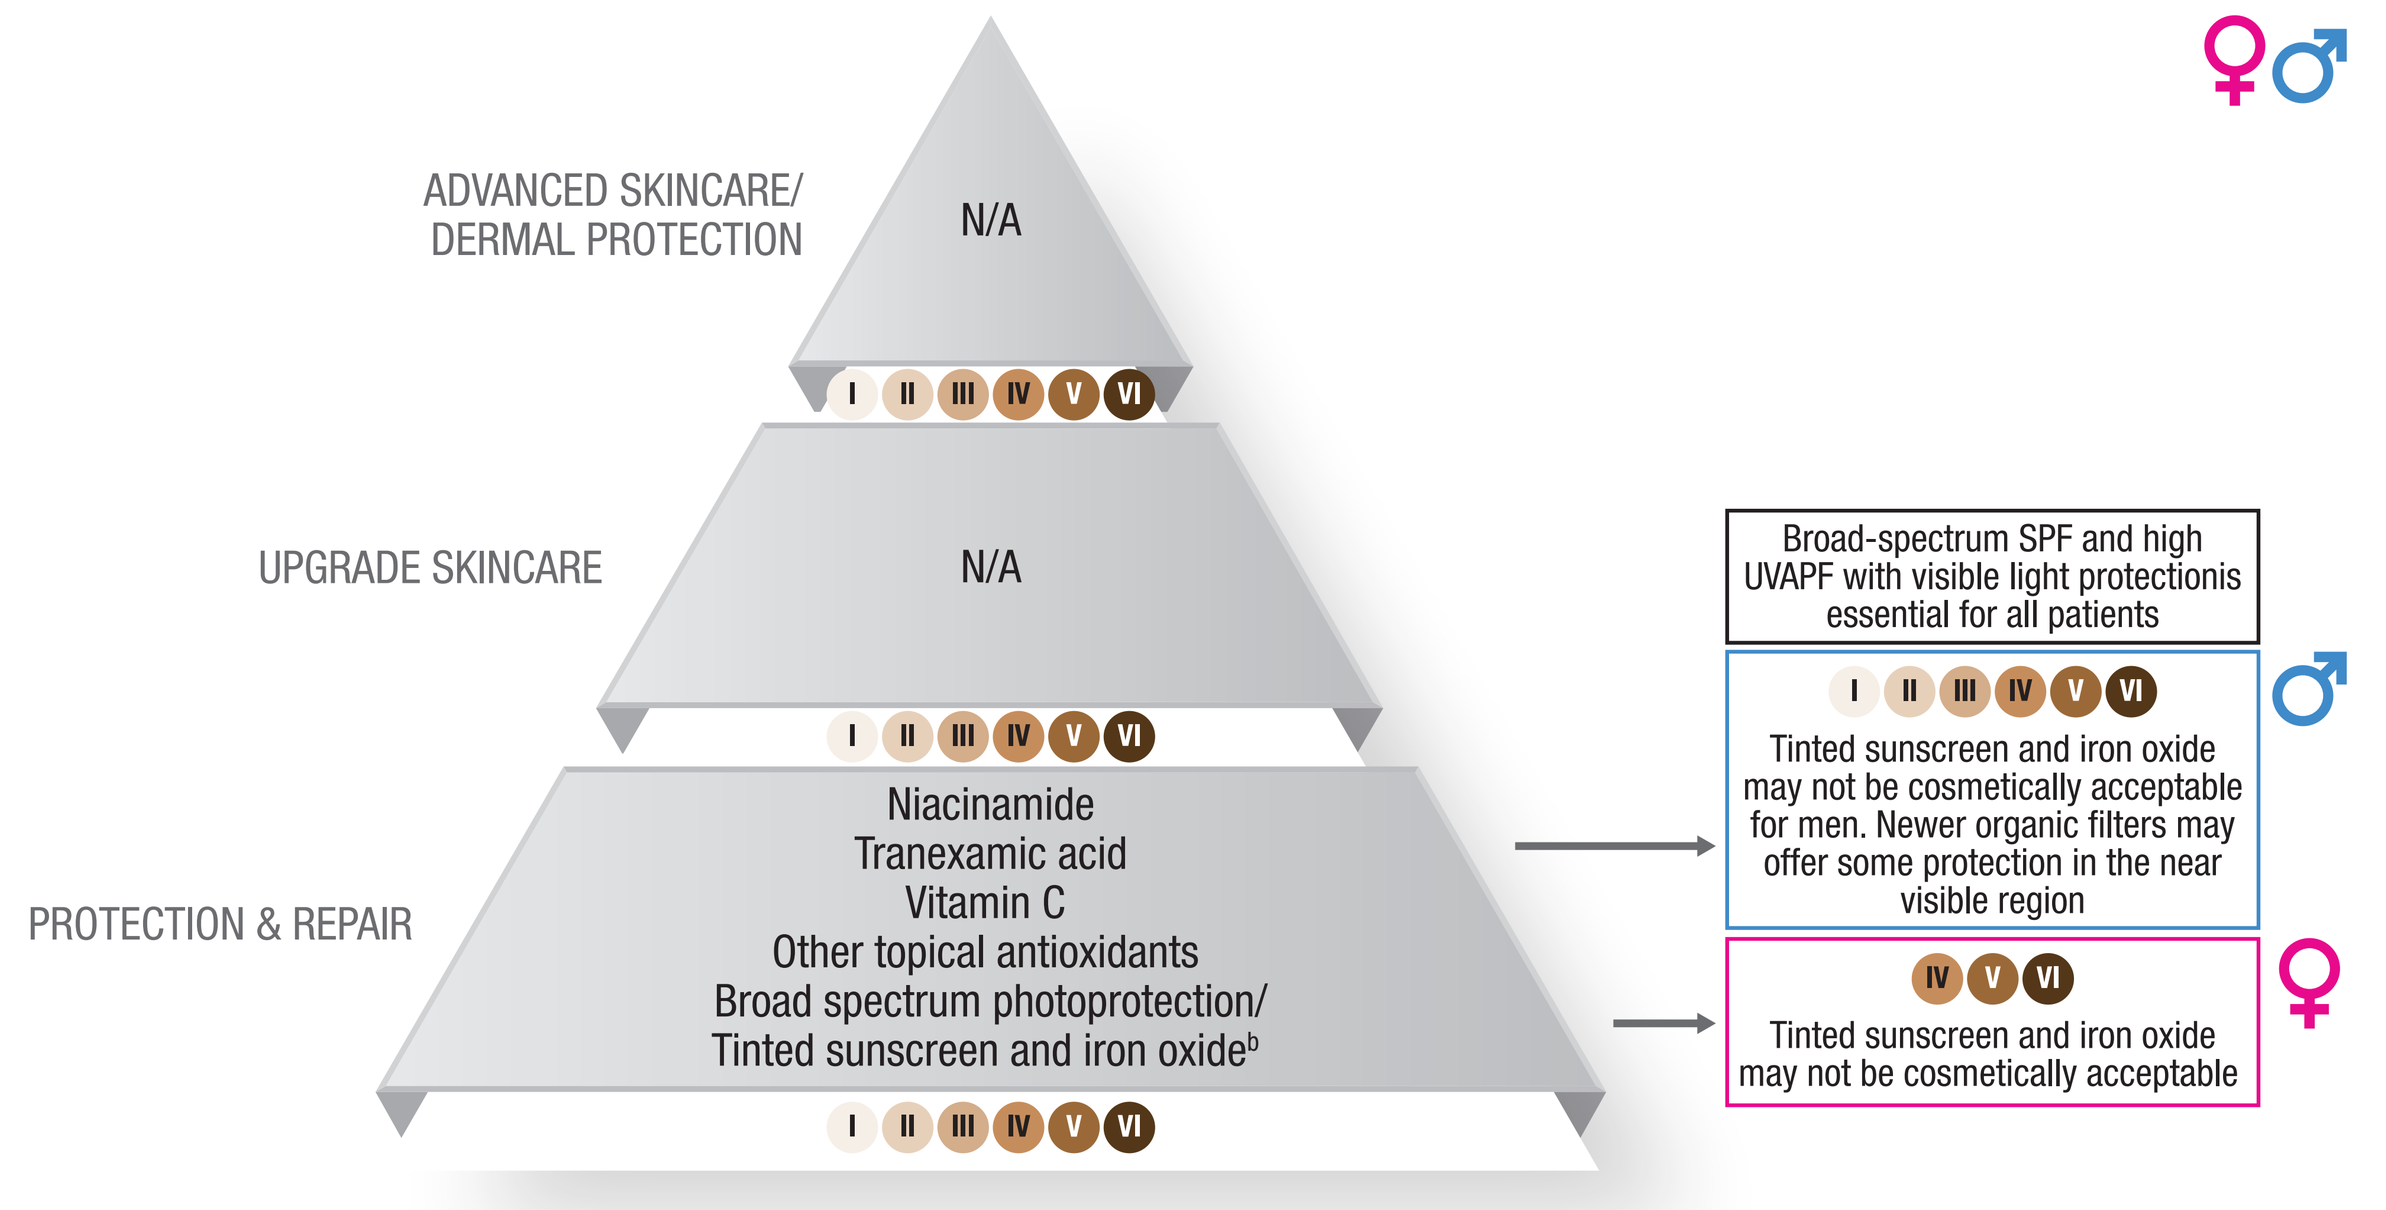
ADVANCED SKINCARE/ DERMAL PROTECTION
N/A
I II III IV V VI
UPGRADE SKINCARE
N/A
I II III IV V VI
PROTECTION & REPAIR
Niacinamide
Tranexamic acid
Vitamin C
Other topical antioxidants
Broad spectrum photoprotection/ Tinted sunscreen and iron oxide[b]
I II III IV V VI
Broad-spectrum SPF and high UVAPF with visible light protectionis essential for all patients
I II III IV V VI
Tinted sunscreen and iron oxide may not be cosmetically acceptable for men. Newer organic filters may offer some protection in the near visible region
IV V VI
Tinted sunscreen and iron oxide may not be cosmetically acceptable

SCENARIO 1

A 30-year-old female FST IV has a 3-month-old son and recently noticed upper lip, bilateral jawline, and lateral forehead pigmentation. She is concerned that the melasma pigmentation continues to darken despite her avoidance of the outdoors. She does not wear photoprotection as she is dissatisfied with the sunscreen appearance on her skin.

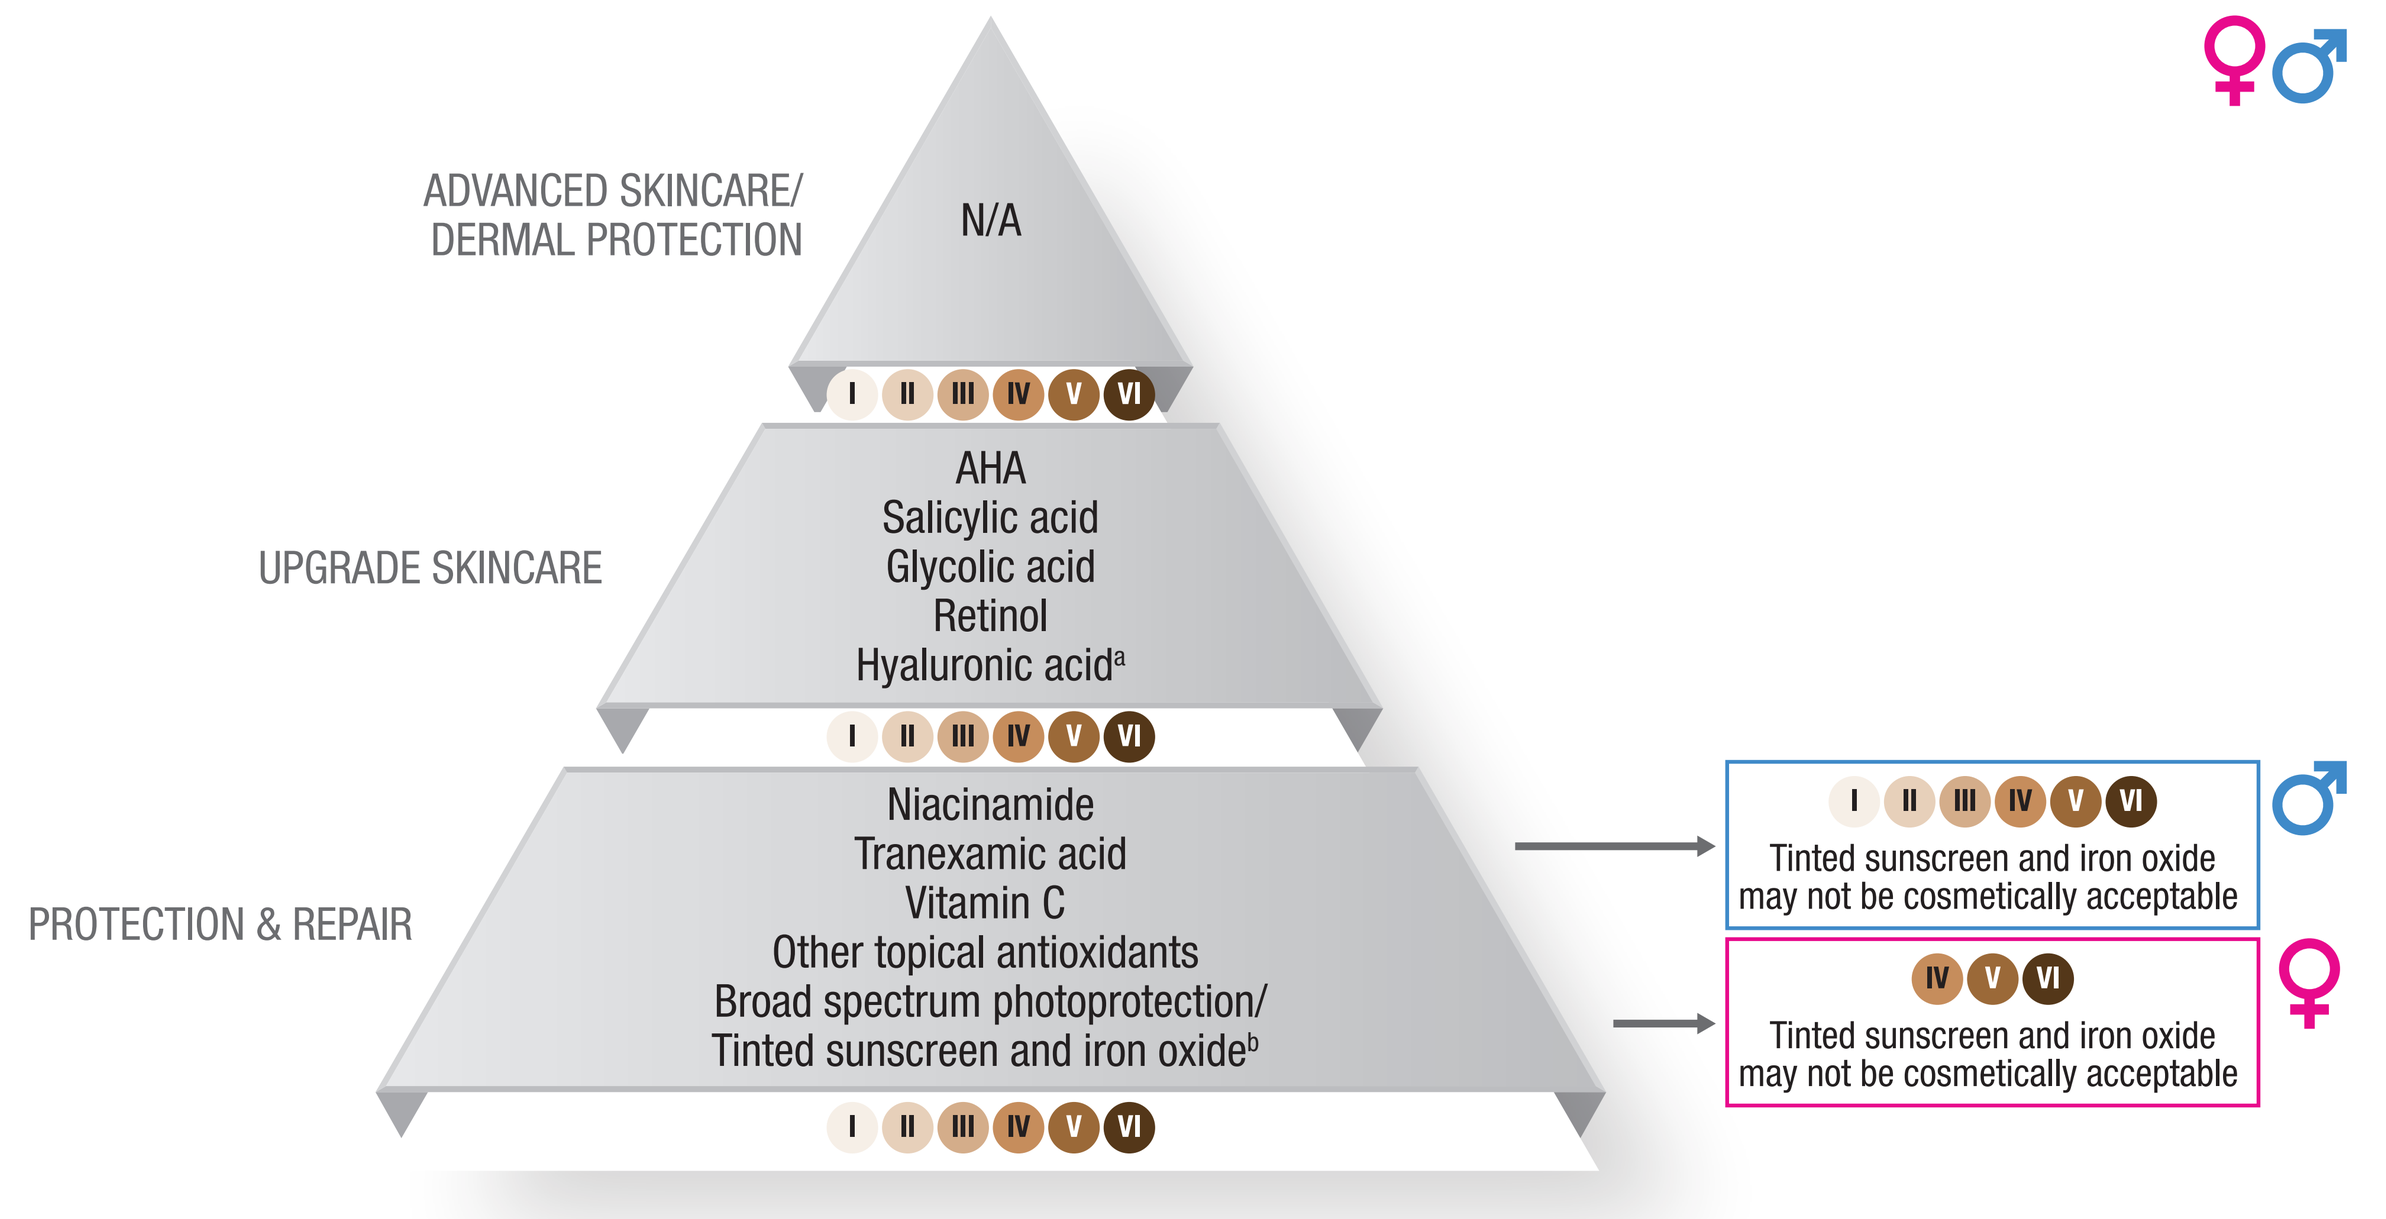
ADVANCED SKINCARE/ DERMAL PROTECTION
N/A
I II III IV V VI
UPGRADE SKINCARE
AHA
Salicylic acid
Glycolic acid
Retinol
Hyaluronic acid[a]
I II III IV V VI
PROTECTION & REPAIR
Niacinamide
Tranexamic acid
Vitamin C
Other topical antioxidants
Broad spectrum photoprotection/ Tinted sunscreen and iron oxide[b]
I II III IV V VI
I II III IV V VI
Tinted sunscreen and iron oxide may not be cosmetically acceptable
IV V VI
Tinted sunscreen and iron oxide may not be cosmetically acceptable

SCENARIO 2

A 25-year-old female with FST III has post-inflammatory hyperpigmentation (PIH) from acne scarring from recent and older lesions. She is noticing the first signs of aging with fine lines around the eyes and desires skin care regimen recommendations for preventing acne and signs of aging.

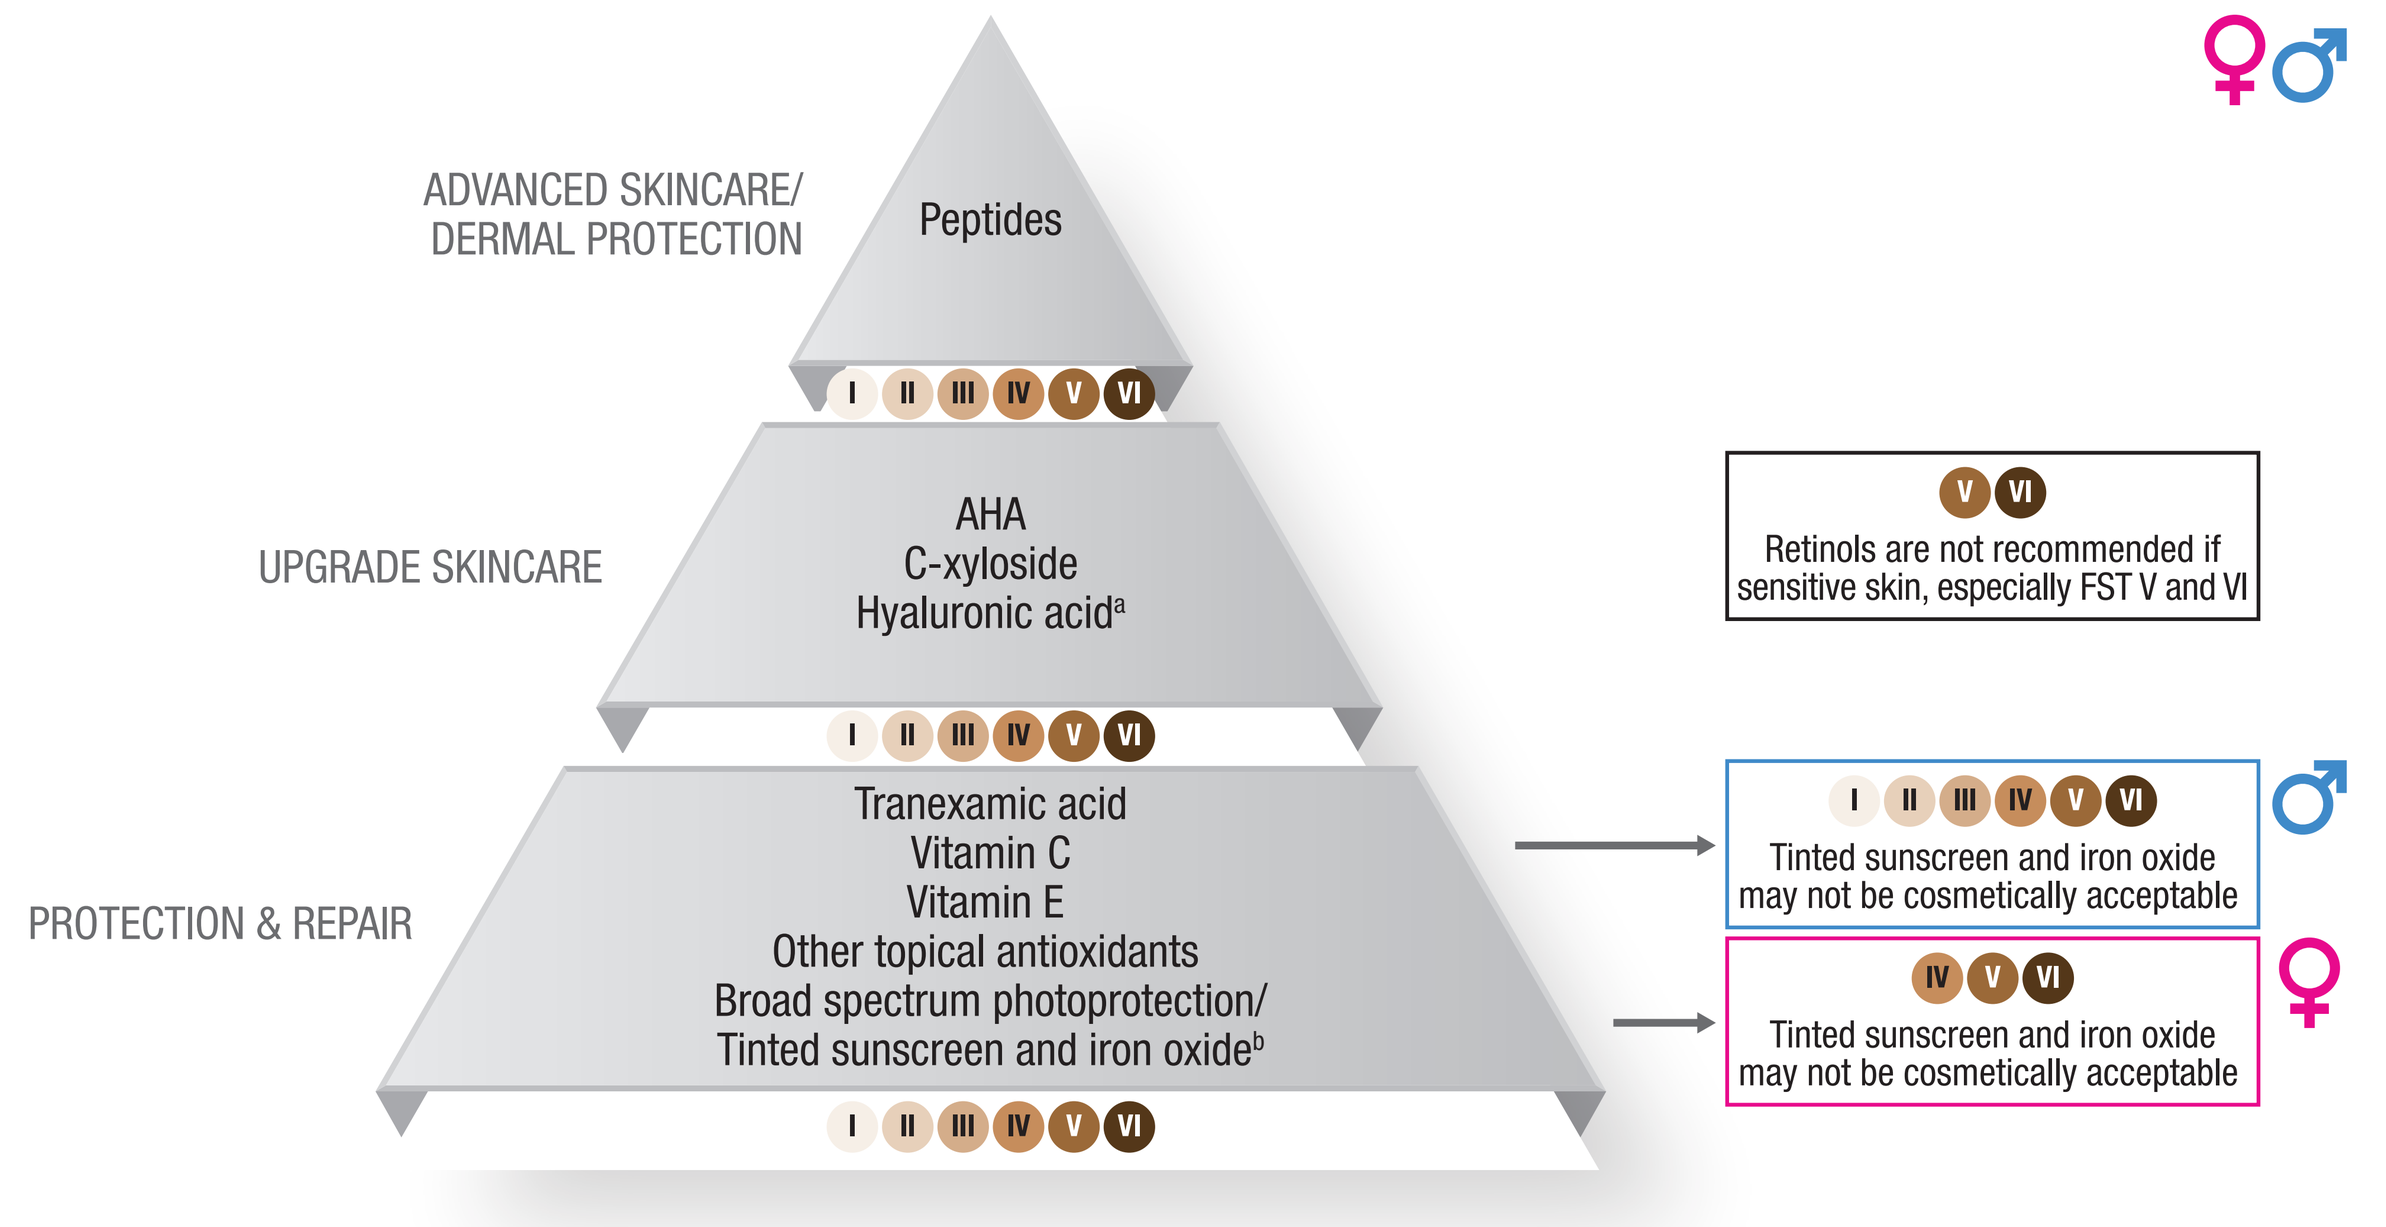
ADVANCED SKINCARE/ DERMAL PROTECTION
Peptides
I II III IV V VI
UPGRADE SKINCARE
AHA
C-xyloside
Hyaluronic acid[a]
I II III IV V VI
PROTECTION & REPAIR
Tranexamic acid
Vitamin C
Vitamin E
Other topical antioxidants
Broad spectrum photoprotection/ Tinted sunscreen and iron oxide[b]
I II III IV V VI
V VI
Retinols are not recommended if sensitive skin, especially FST V and VI
I II III IV V VI
Tinted sunscreen and iron oxide may not be cosmetically acceptable
IV V VI
Tinted sunscreen and iron oxide may not be cosmetically acceptable

SCENARIO 3

A 30-year-old FST II female has sensitive skin and desires to initiate anti-aging cosmetic solutions for the first mild signs of aging as she has lentigines on her face, fine glabellar lines and facial dryness. She has a 6-month-old daughter so frequently lacks sleep and she works outdoors.

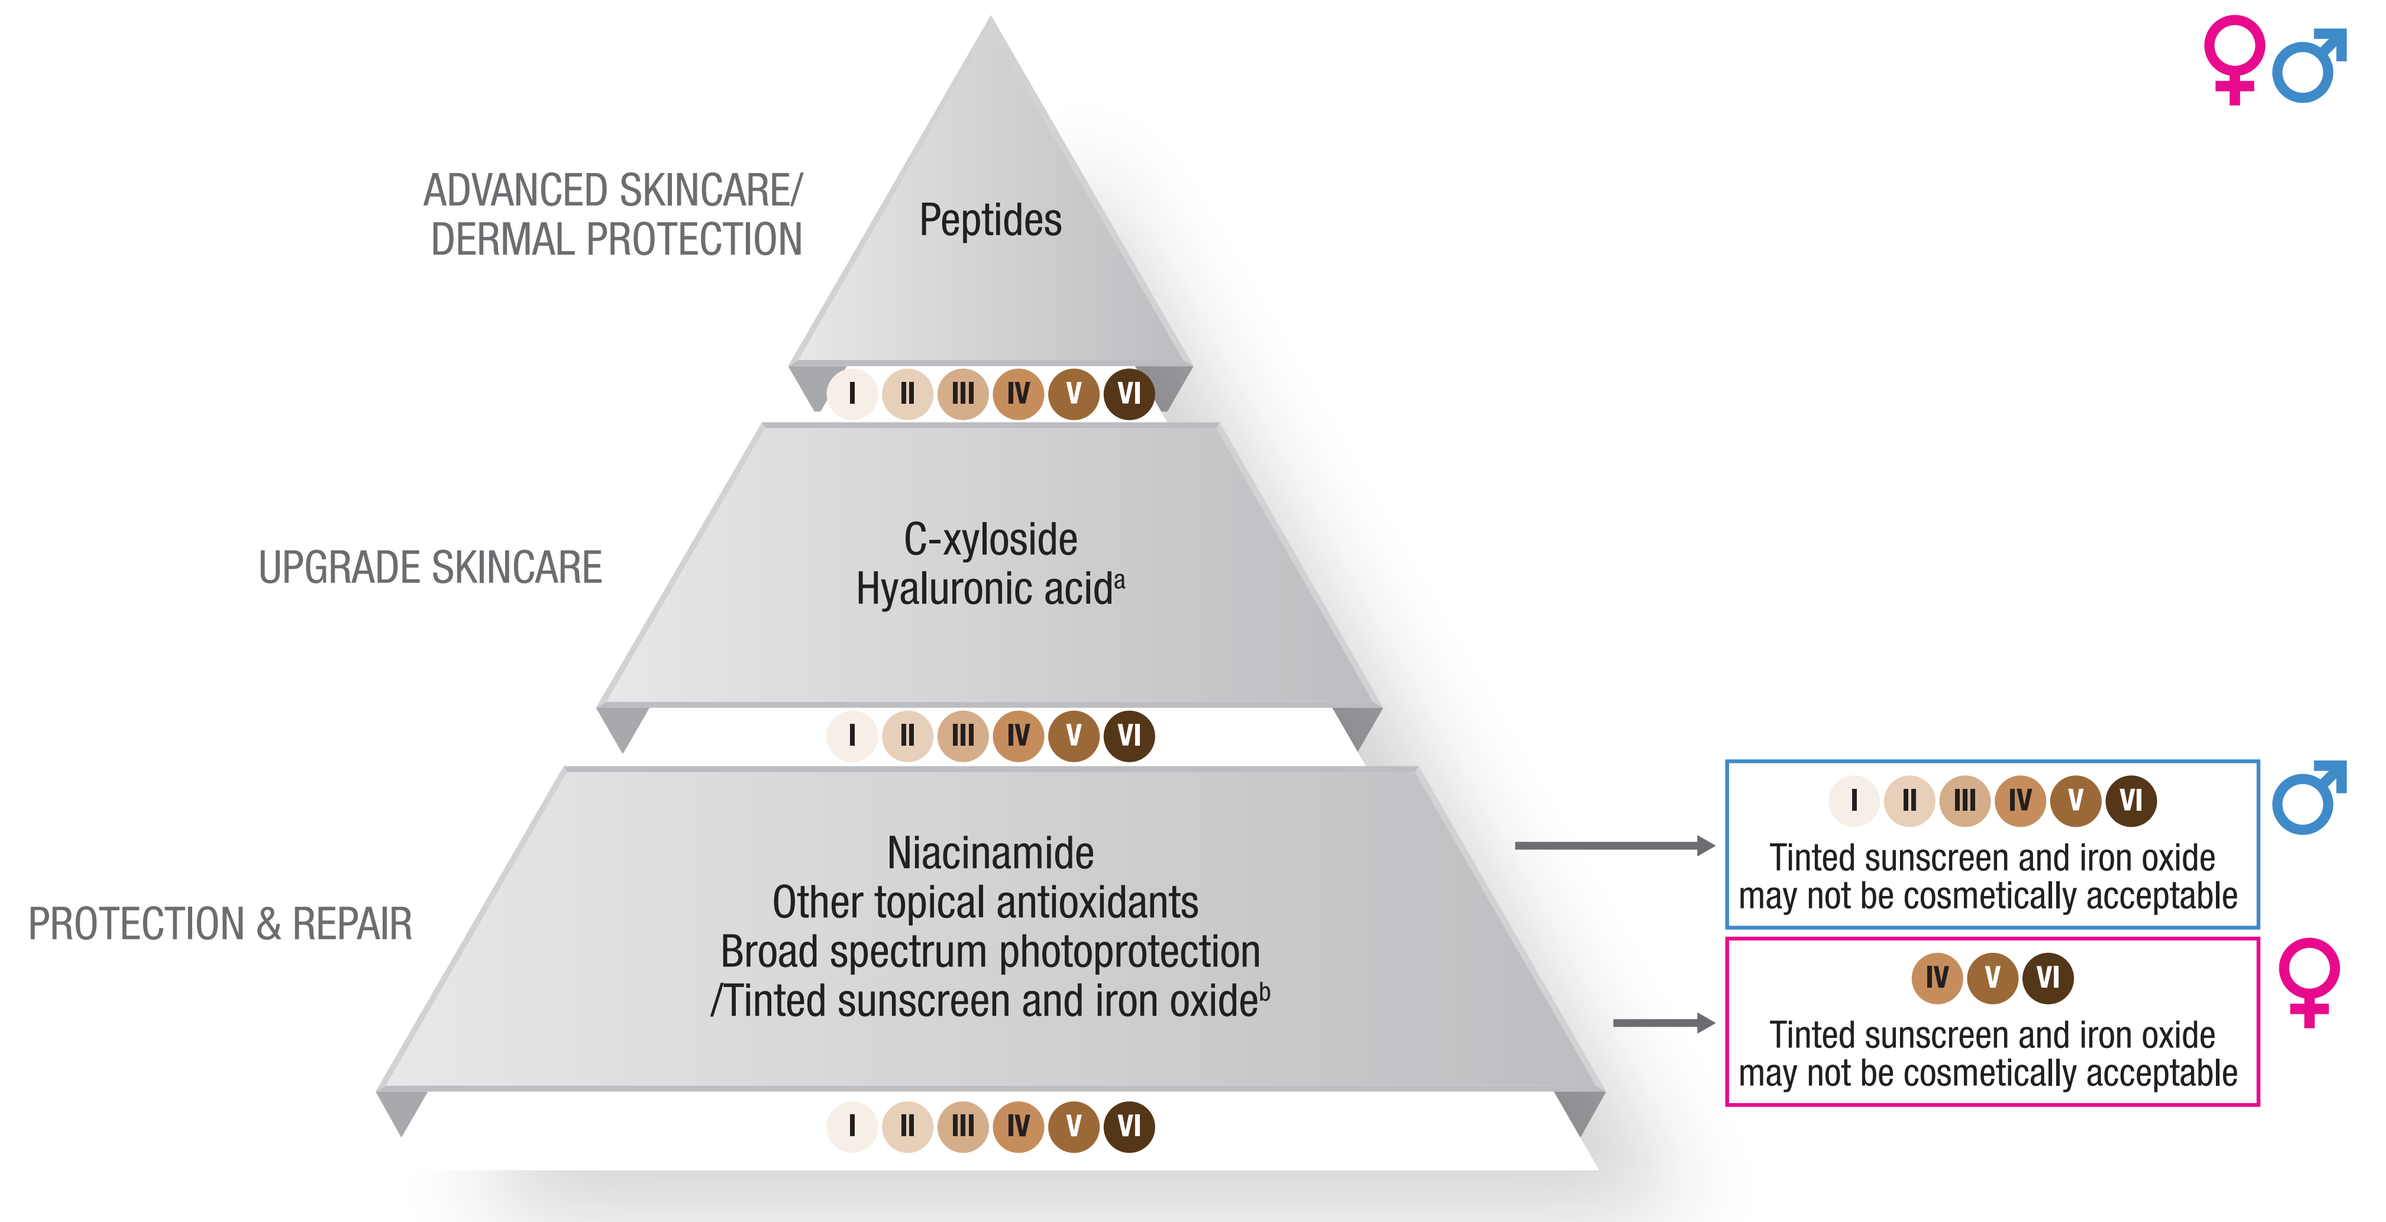
ADVANCED SKINCARE/ DERMAL PROTECTION
Peptides
I II III IV V VI
UPGRADE SKINCARE
C-xyloside
Hyaluronic acid[a]
I II III IV V VI
PROTECTION & REPAIR
Niacinamide
Other topical antioxidants
Broad spectrum photoprotection /Tinted sunscreen and iron oxide[b]
I II III IV V VI
I II III IV V VI
Tinted sunscreen and iron oxide may not be cosmetically acceptable
IV V VI
Tinted sunscreen and iron oxide may not be cosmetically acceptable

SCENARIO 4

A 35-year-old FST III male wants to both improve his appearance and prevent photoaging.
He completed a course of oral isotretinoin 3 months ago for treating his cystic acne and is noticing a few isolated papules and pustules and xerosis.
He wishes to resume sky diving, but has photosensitivity from the oral retinoid.

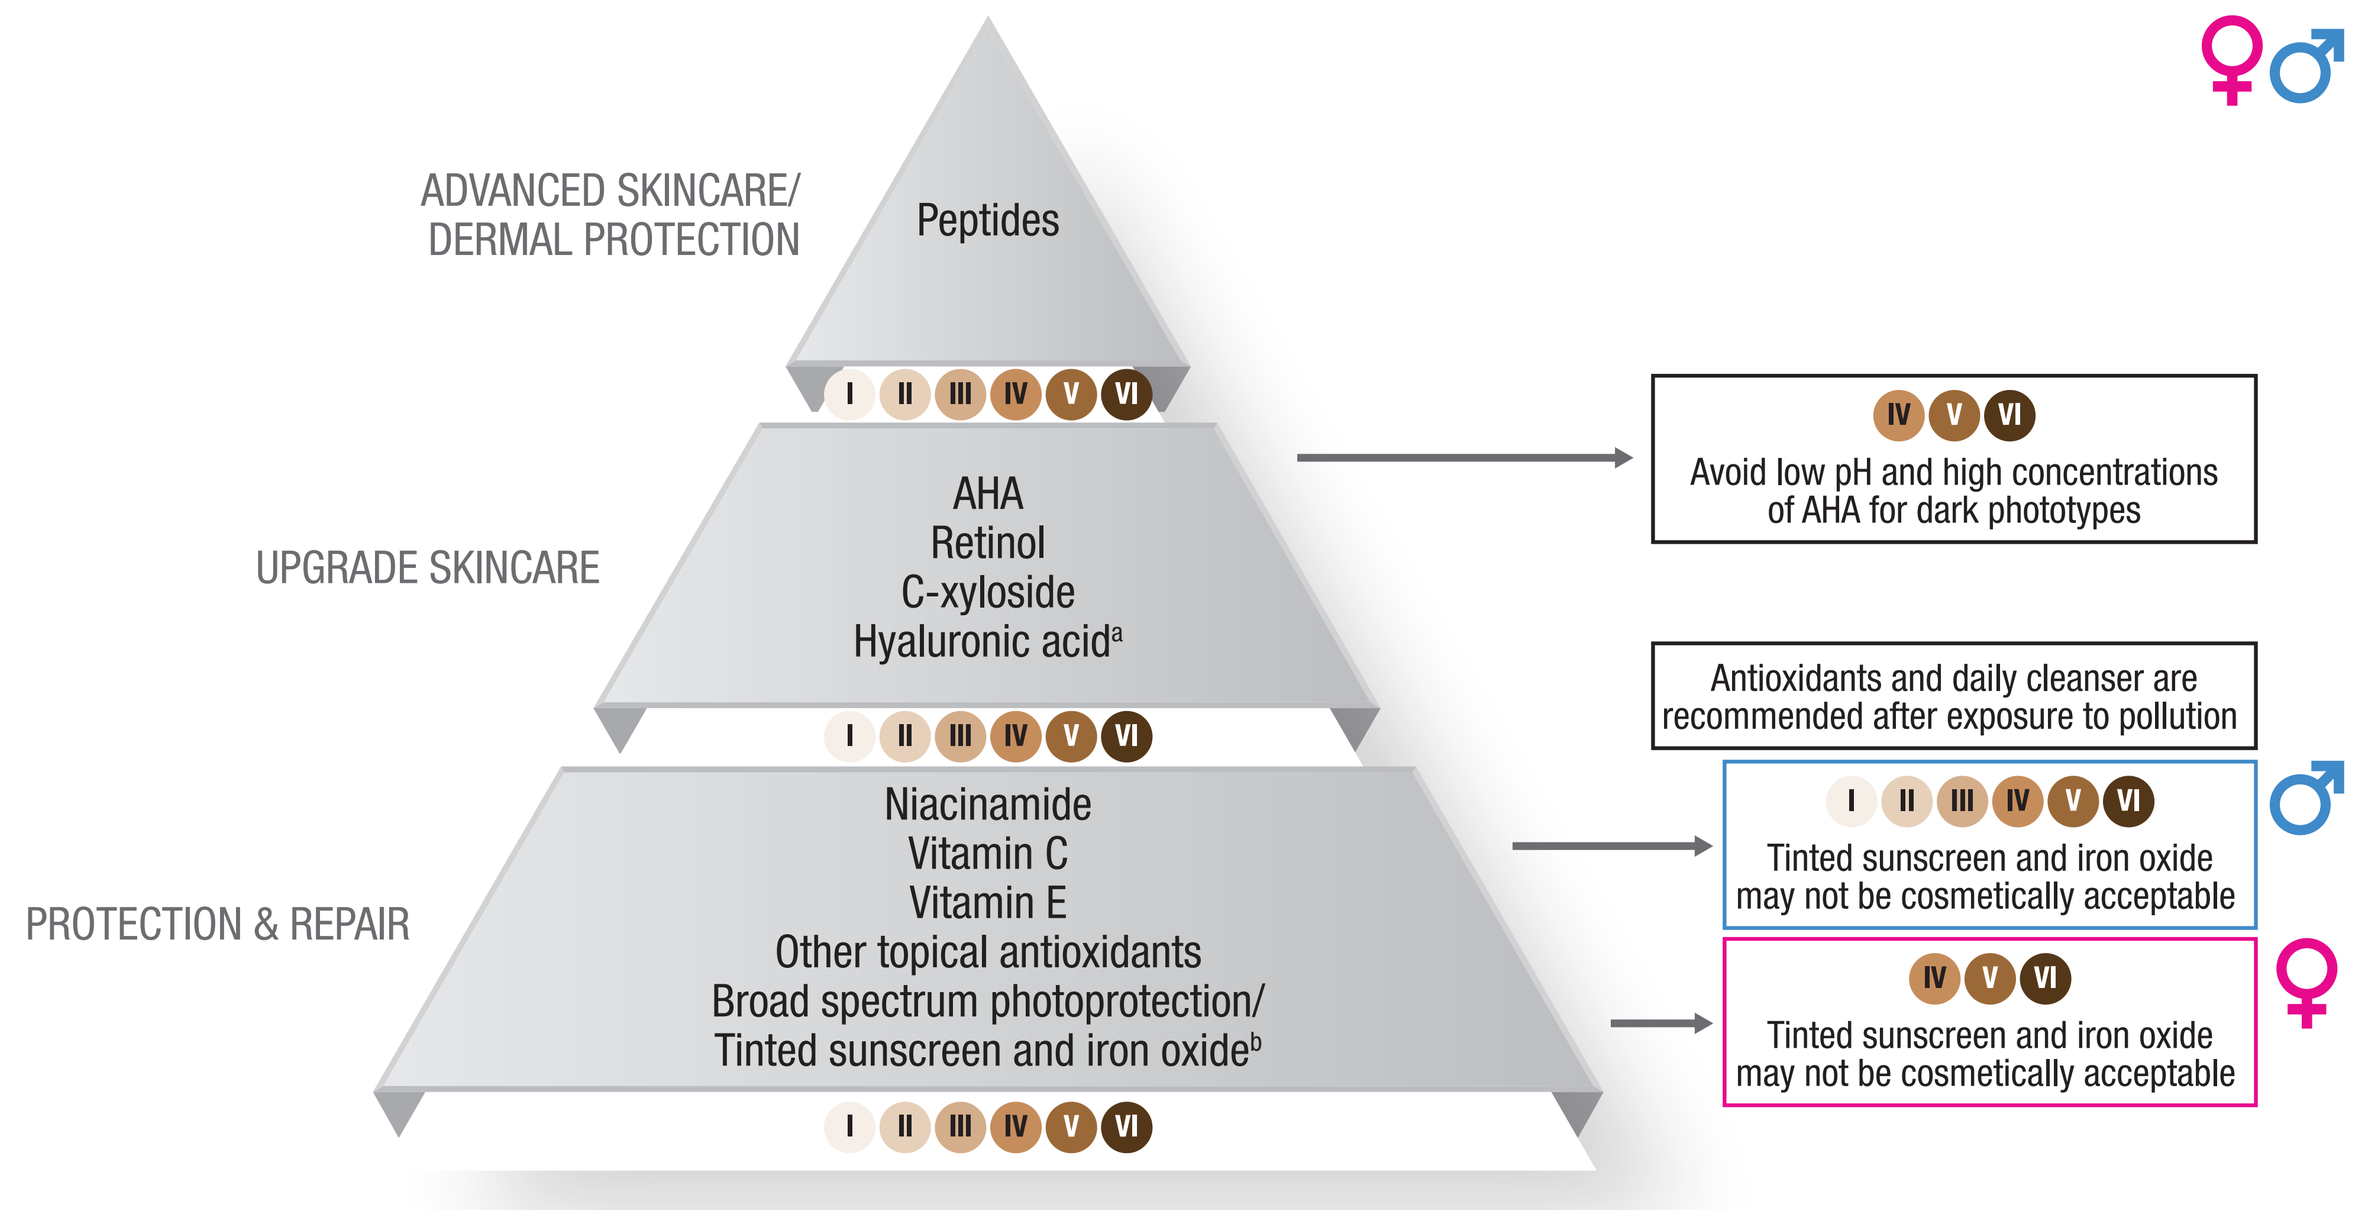
ADVANCED SKINCARE/ DERMAL PROTECTION
Peptides
I II III IV V VI
UPGRADE SKINCARE
AHA
Retinol
C-xyloside
Hyaluronic acid[a]
I II III IV V VI
PROTECTION & REPAIR
Niacinamide
Vitamin C
Vitamin E
Other topical antioxidants
Broad spectrum photoprotection/ Tinted sunscreen and iron oxide[b]
I II III IV V VI
IV V VI
Avoid low pH and high concentrations of AHA for dark phototypes
Antioxidants and daily cleanser are recommended after exposure to pollution
I II III IV V VI
Tinted sunscreen and iron oxide may not be cosmetically acceptable
IV V VI
Tinted sunscreen and iron oxide may not be cosmetically acceptable

SCENARIO 5

A 40-year-old female with FST I has initiated botulinum toxin treatment for her glabellar lines and had hyaluronic acid injected into her nasolabial folds.
She wants dermocosmetic solutions to complement the antiaging procedures and optimize her skin texture. She always uses sunscreen but lives and works in an area exposed to urban air pollution.

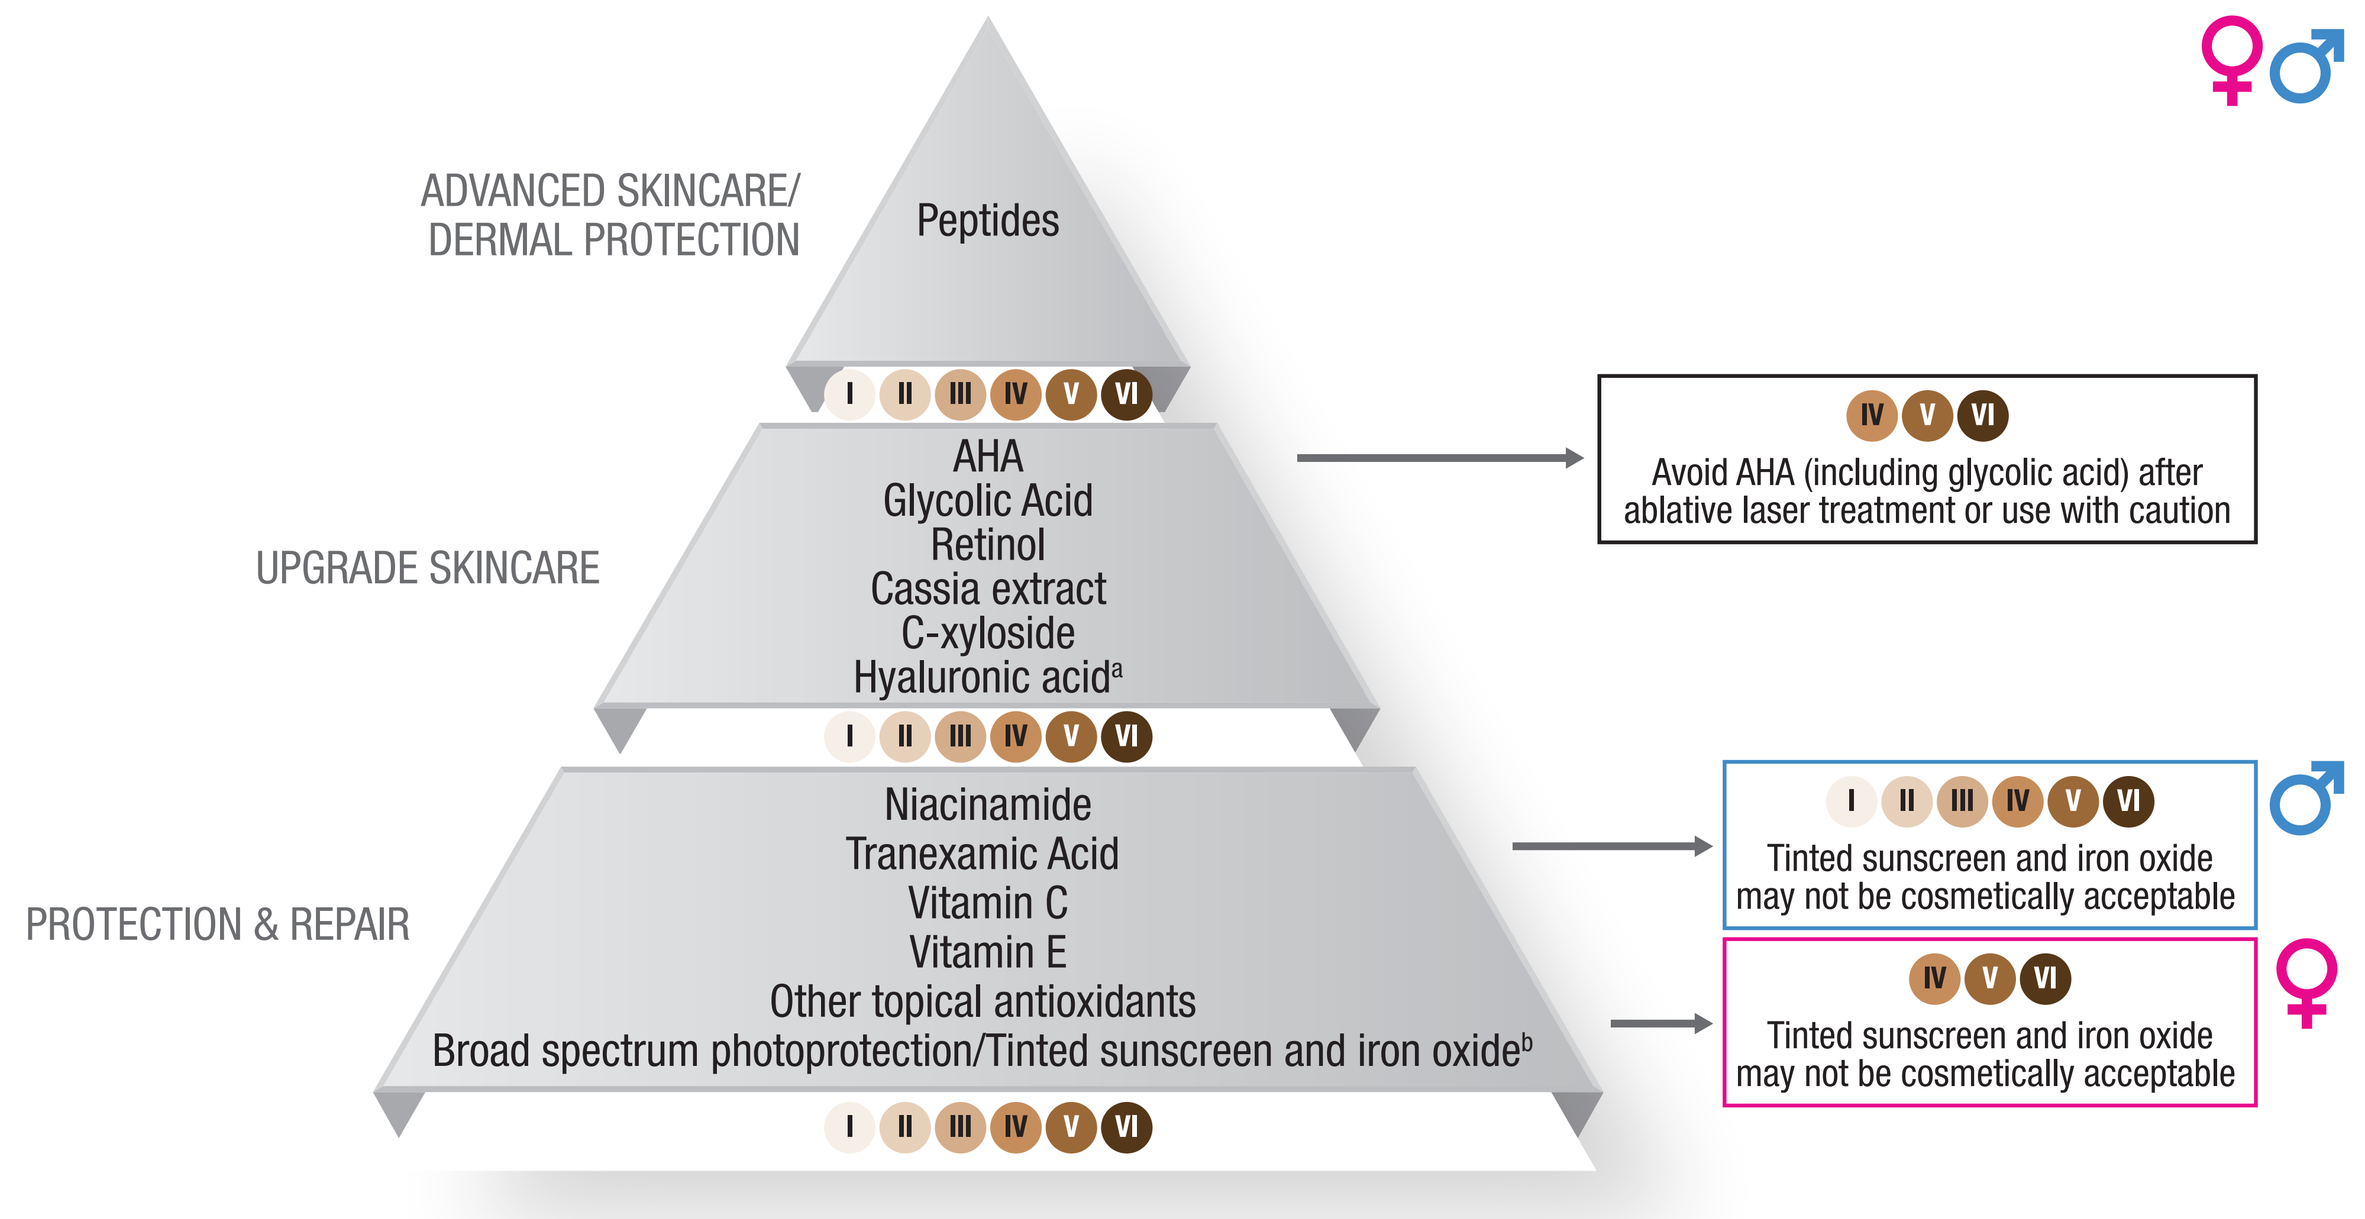
ADVANCED SKINCARE/ DERMAL PROTECTION
Peptides
I II III IV V VI
UPGRADE SKINCARE
AHA
Glycolic Acid
Retinol
Cassia extract
C-xyloside
Hyaluronic acid[a]
I II III IV V VI
PROTECTION & REPAIR
Niacinamide
Tranexamic Acid
Vitamin C
Vitamin E
Other topical antioxidants
Broad spectrum photoprotection/Tinted sunscreen and iron oxide[b]
I II III IV V VI
IV V VI
Avoid AHA (including glycolic acid) after ablative laser treatment or use with caution
I II III IV V VI
Tinted sunscreen and iron oxide may not be cosmetically acceptable
IV V VI
Tinted sunscreen and iron oxide may not be cosmetically acceptable

SCENARIO 6

A 45-year-old female with FST II and pigmentation wishes to improve her skin performance. She has tried various dermocosmetics without the rapid results she desires. She elected to have a carbon dioxide laser resurfacing procedure and wants recommendations for both pre- and post-procedure skincare.

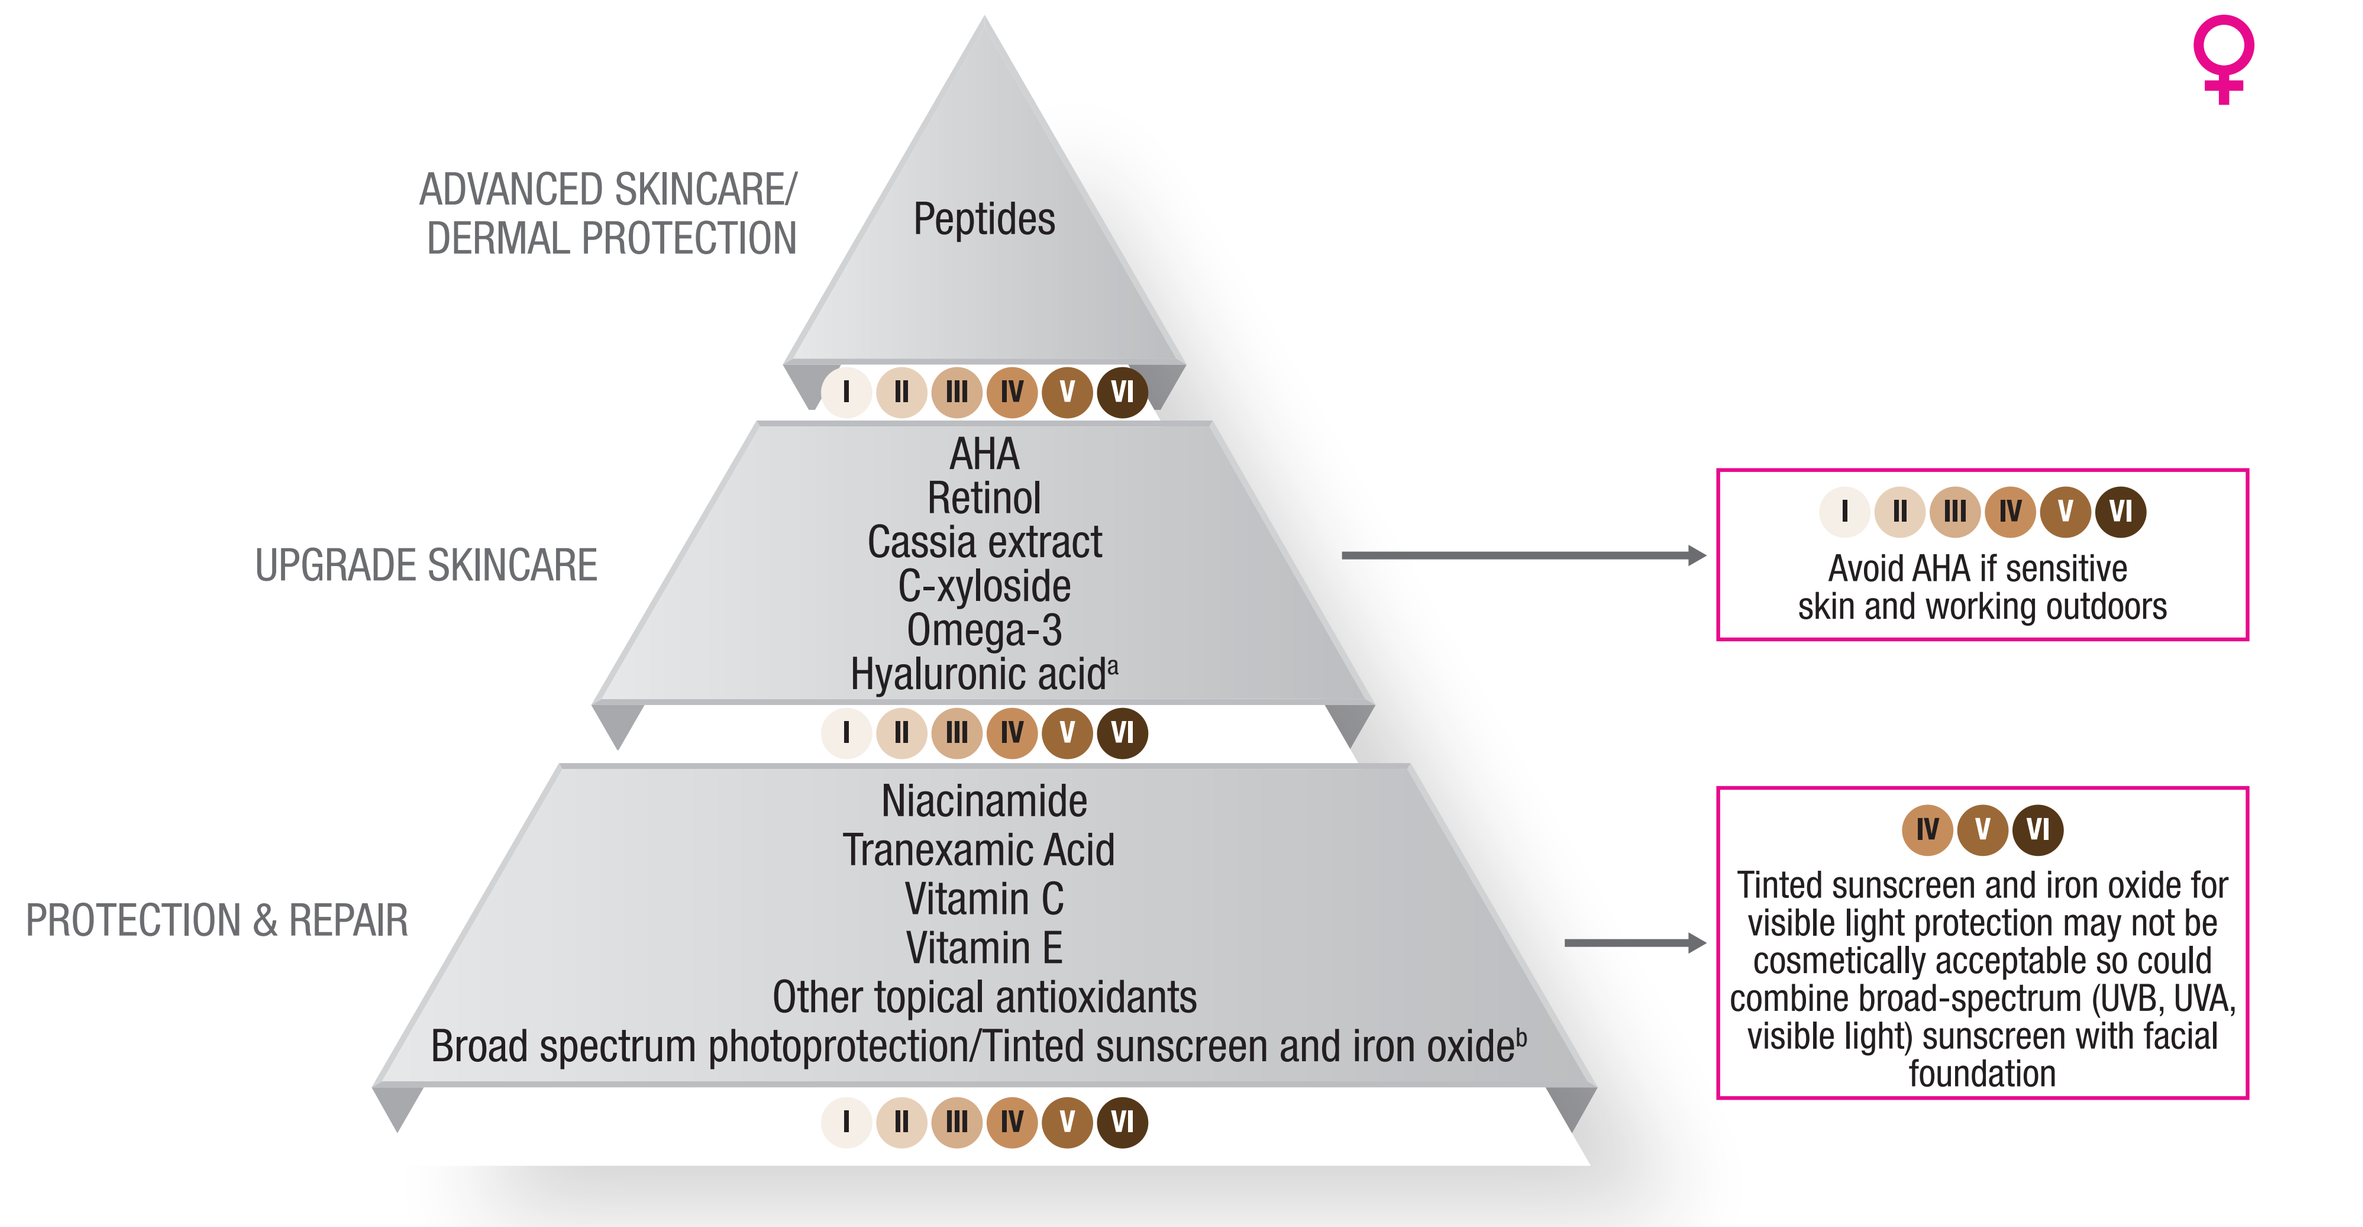
ADVANCED SKINCARE/ DERMAL PROTECTION
Peptides
I II III IV V VI
UPGRADE SKINCARE
AHA
Retinol
Cassia extract
C-xyloside
Omega-3
Hyaluronic acid[a]
I II III IV V VI
PROTECTION & REPAIR
Niacinamide
Tranexamic Acid
Vitamin C
Vitamin E
Other topical antioxidants
Broad spectrum photoprotection/Tinted sunscreen and iron oxide[b]
I II III IV V VI
I II III IV V VI
Avoid AHA if sensitive skin and working outdoors
IV V VI
Tinted sunscreen and iron oxide for visible light protection may not be cosmetically acceptable so could combine broad-spectrum (UVB, UVA, visible light) sunscreen with facial foundation

SCENARIO 7

A 45-year-old peri-menopausal female with FST IV has just started working as a live on-location television reporter. With the increased sun exposure, she is noticing actinic pigmentation on her bilateral cheeks and wants recommendations for sunscreen that will not appear white and pasty on her skin. She has hot flashes and her skin aspect is changing so she no longer tolerates her usual cosmetic routine.

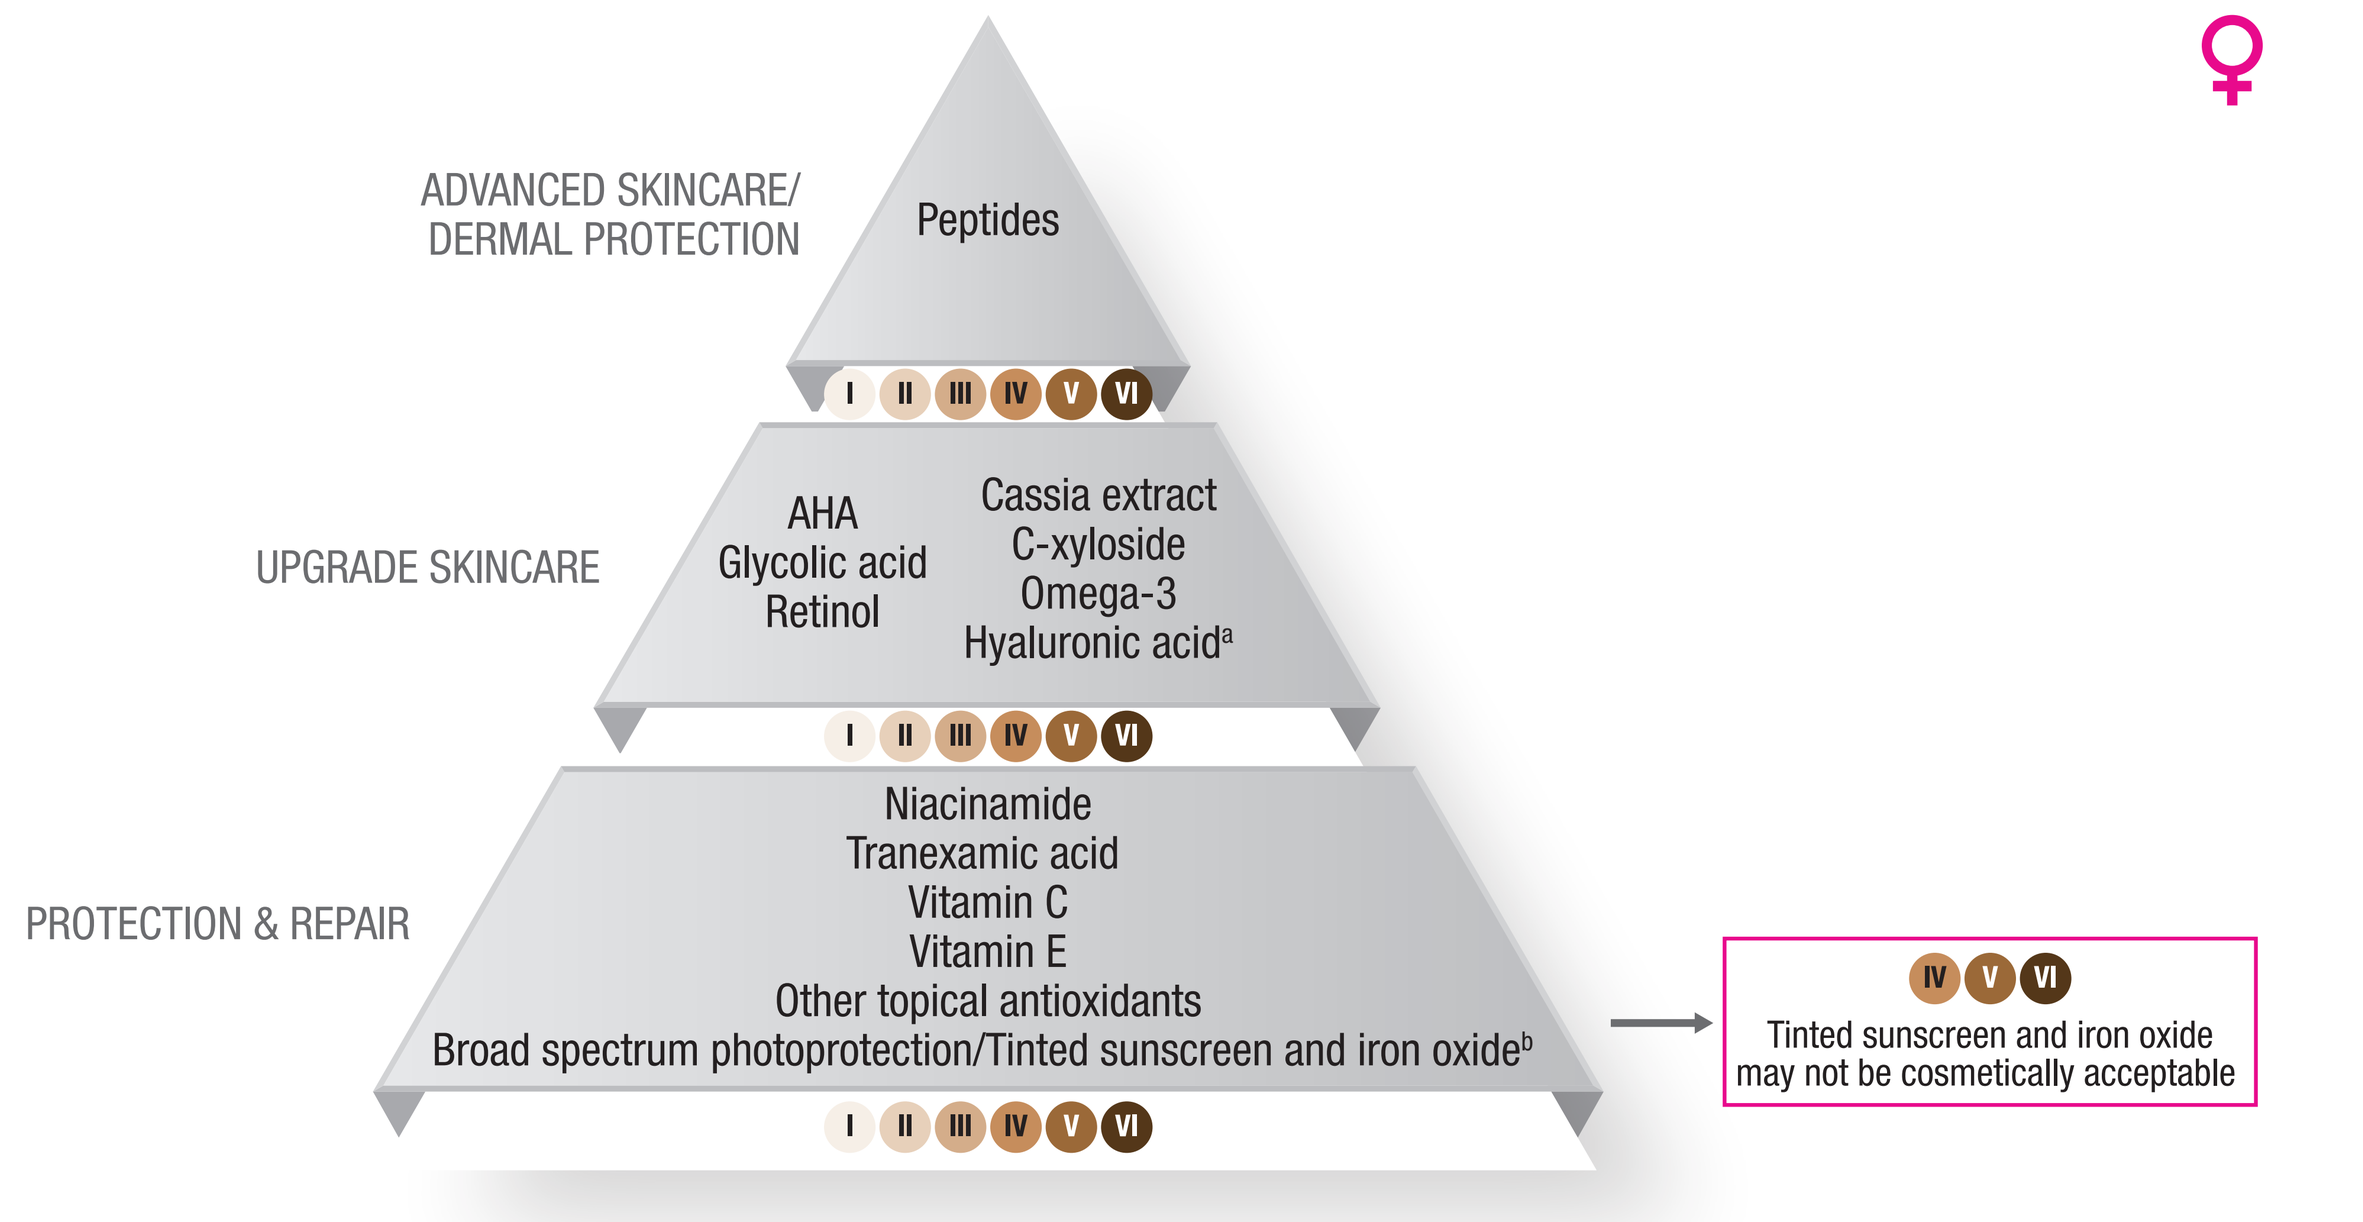
ADVANCED SKINCARE/ DERMAL PROTECTION
Peptides
I II III IV V VI
UPGRADE SKINCARE
AHA
Glycolic acid
Retinol
Cassia extract
C-xyloside
Omega-3
Hyaluronic acid[a]
I II III IV V VI
PROTECTION & REPAIR
Niacinamide
Tranexamic acid
Vitamin C
Vitamin E
Other topical antioxidants
Broad spectrum photoprotection/Tinted sunscreen and iron oxide[b]
I II III IV V VI
IV V VI
Tinted sunscreen and iron oxide may not be cosmetically acceptable

SCENARIO 8

A 60-year-old menopausal female with FST I desires suggestions to improve her appearance.
She notes upper lip and jawline dyspigmentation that has worsened considerably since she began estrogen replacement therapy. Until her recent retirement, she was a heavy smoker due to the stress of her job.

4 DISCUSSION

- We describe a simple, practical tool for use in daily dermatology consultations that is adapted to specific patient needs.
- This work provides recommendations to cover diverse and inclusive populations of patients, addressing all skin types and international needs.

Abbreviations: AHA alpha hydroxy acids; FST Fitzpatrick skin type; [a]Low-high molecular weight hyaluronic acid; [b]Broad-spectrum photoprotection with high SPF + UVAPF and tinted sunscreen/iron oxide visible light protection.

References: 1. Mayoral FA, Kenner JR, Draelos ZD. The skin health and beauty pyramid: a clinically based guide to selecting topical skincare products. J Drugs Dermatol. Apr 2014;13(4):414-21. 2. Draelos ZD. Revisiting the Skin Health and Beauty Pyramid: A Clinically Based Guide to Selecting Topical Skincare Products. J Drugs Dermatol. Jun 1 2021;20(6):695-699. doi:10.36849/jdd.2021.5883.

Acknowledgements: Study and poster/editorial support funded by Vichy (L'Oréal).