EVALUATION OF A DERMOCOSMETIC IN SUBJECTS WITH AN ALLERGIC DIATHESIS AND INTOLERANT TO COSMETIC SKIN CARE

Jerry Tan[1], Ann'Laure Demessant[2], Guénaëlle Le Dantec[2] Caroline le Floc'h[2], Delphine Kerob[2]

[1]Western University, Department of Medicine and Windsor Clinical Research Inc, Windsor, ON, Canada, [2]La Roche-Posay Laboratoire Dermatologique, Levallois-Perret, France

INTRODUCTION

Intolerance to cosmetics and dermocosmetics is a frequent issue seen in patients with allergic contact dermatitis (ACD) and sensitive skin.[1-3] In allergic patients the skin barrier is altered, and the skin microbiome is unbalanced.[4, 5]

Allergic and immunologic skin diseases maynegatively impact the quality of life (QoL) of affected patients with detrimental consequences.[6] The tested dermocosmetic (DC) has been specifically developed to restore the natural skin barrier and rebalance the skin immune system to relief from signs and symptoms related to skin intolerance. Key ingredients are Niacinamide, fractions of the probiotic Sphingobioma and Neurosensine, a soothing compound that acts on skin sensitivity in decreasing erythema, irritation and pruritus which all have been described in patients with ACD.[7]

This study assessed the local tolerance and efficacy of a specifically developed DC in subjects with an allergic background and intolerance to dermocosmetics.

MATERIAL & METHODS

This was an open-labeled, multicenter study conducted under dermatological control in Caucasian subjects above 16 years of age with an allergic background and intolerance to cosmetics since at least 2 years prior to inclusion. The DC was to be applied on the entire face twice daily for 28 days.

Dermatological assessments at D0, D14 and D28 included the composite skin sensitivity score(sum of pruritus, redness, burning sensation and tingling on a scale from 0=non to 4=severe) and symptoms (pruritus, redness, burning sensation and tingling), stinging test using the global cutaneous reactivity score, the global Sensiscore assessing the subject-reported facial skin intolerance and reactivity on a scale from 0=never to 4=all the time, local tolerance, transepidermal water loss (TEWL), and skin hydration measured by corneometry. Subjects rated the soothing effect and their satisfaction with the DC, for both face and periocular use.

RESULTS

Of the 107 subjects, 88% were women. The mean age was 42.0±15.0 years. The mean composite score at baseline was 5.9±0.35 (on a scale from 0=absent to 12=severe). The mean global cutaneous reactivity score was 3.9±0.3 at baseline. In total, 53% of the subjects had dry skin, 60% had allergic rhinitis, 46% ACD, 15% allergic conjunctivitis. 95% reported skin prone to irritation, 92% facial sensitive skin and 74% reactive skin (with redness and/or burning, stinging, itching sensation); 88% of the subjects reported reactivity to several cosmetics. Stinging test scores (range: 0-9) significantly decreased from 3.9±0.3 at baseline to 2.4±0.4 at Day 14 (-39%) and 1.4±0.3 (-64%) at Day 28. Overall, 77% and 81% of subjects reported improved skin reactivity at Day 14 and Day 28, respectively. The composite skin sensitivity score decreased from 5.9±0.35 to 0.05±0.1 after 28 days (p<0.0001; Figure 1); so did individual subject-reported symptoms (Figure 2). Significant improvements were noted in the frequency and intensity of signs and symptoms such as skin irritation, erythema, stinging, burning, discomfort. Objective measures showed significant improvements in TEWL (Figure 3) and skin hydration (corneometry, Figure 4) at Day 14 and Day 28. The percentage of subjects with a decreased TEWL was 82% after 14 and 99% after 28 days; 77% of subjects at Day 14 and 97% at Day 28 had an improved skin hydration. Subject satisfaction was high, and tolerance rated good to very good in more than 95% of subjects.

Figure 1

COMPOSITE SKIN SENSITIVITY SCORE AT DAY 0, DAY 14 AND DAY 28

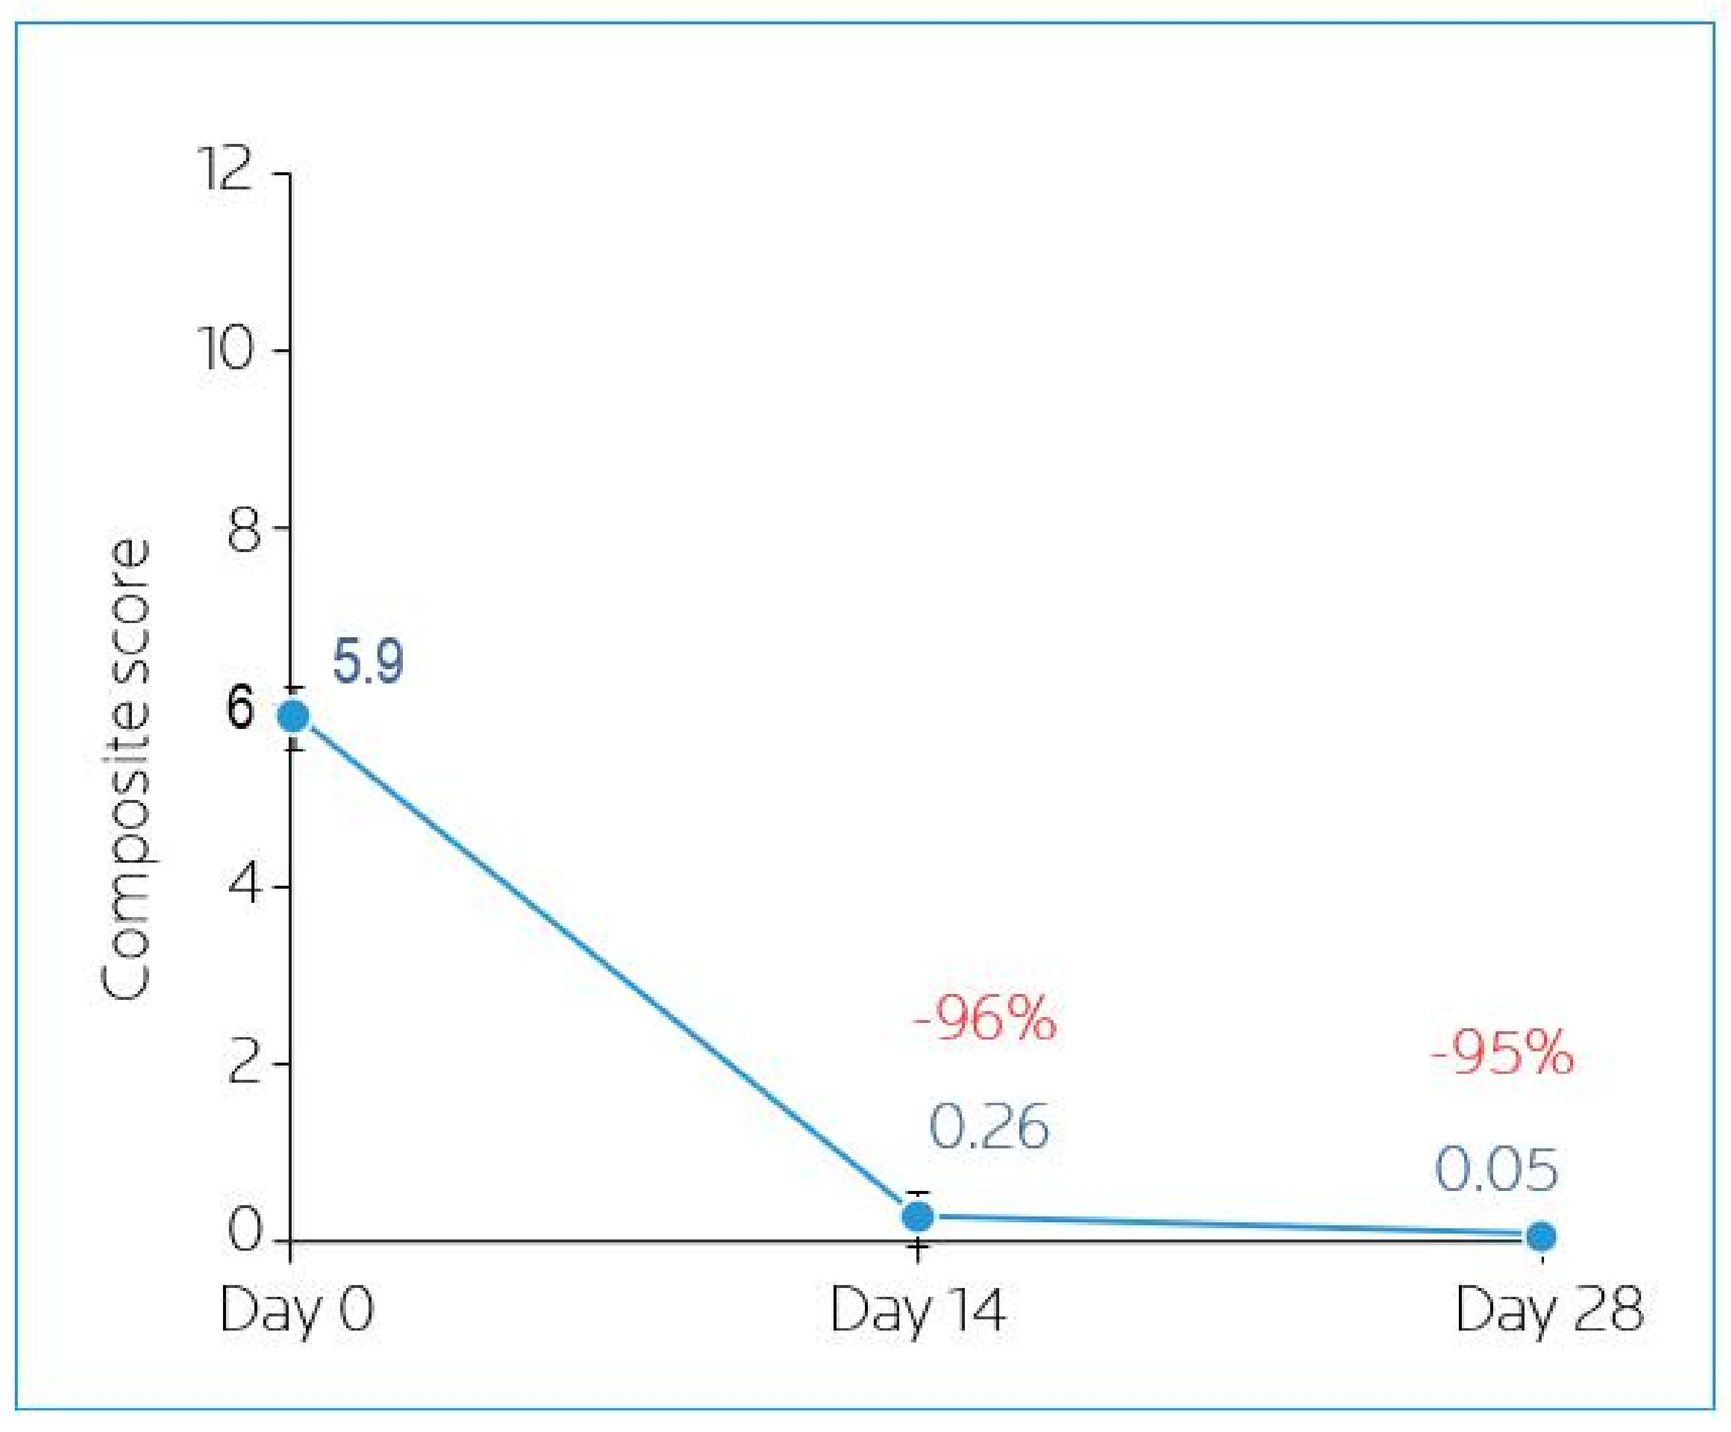
Composite score
12
10
8
6
4
2
0
5.9
-96%
0.26
-95%
0.05
Day 0
Day 14
Day 28

The composite skin sensitivity score significantly (p<0.0001) had decreased at Day 14 and Day 28 compared to Day 0.

Figure 2

SUBJECT-REPORTED SYMPTOM SCORES AT DAY 0, DAY 14 AND DAY 28

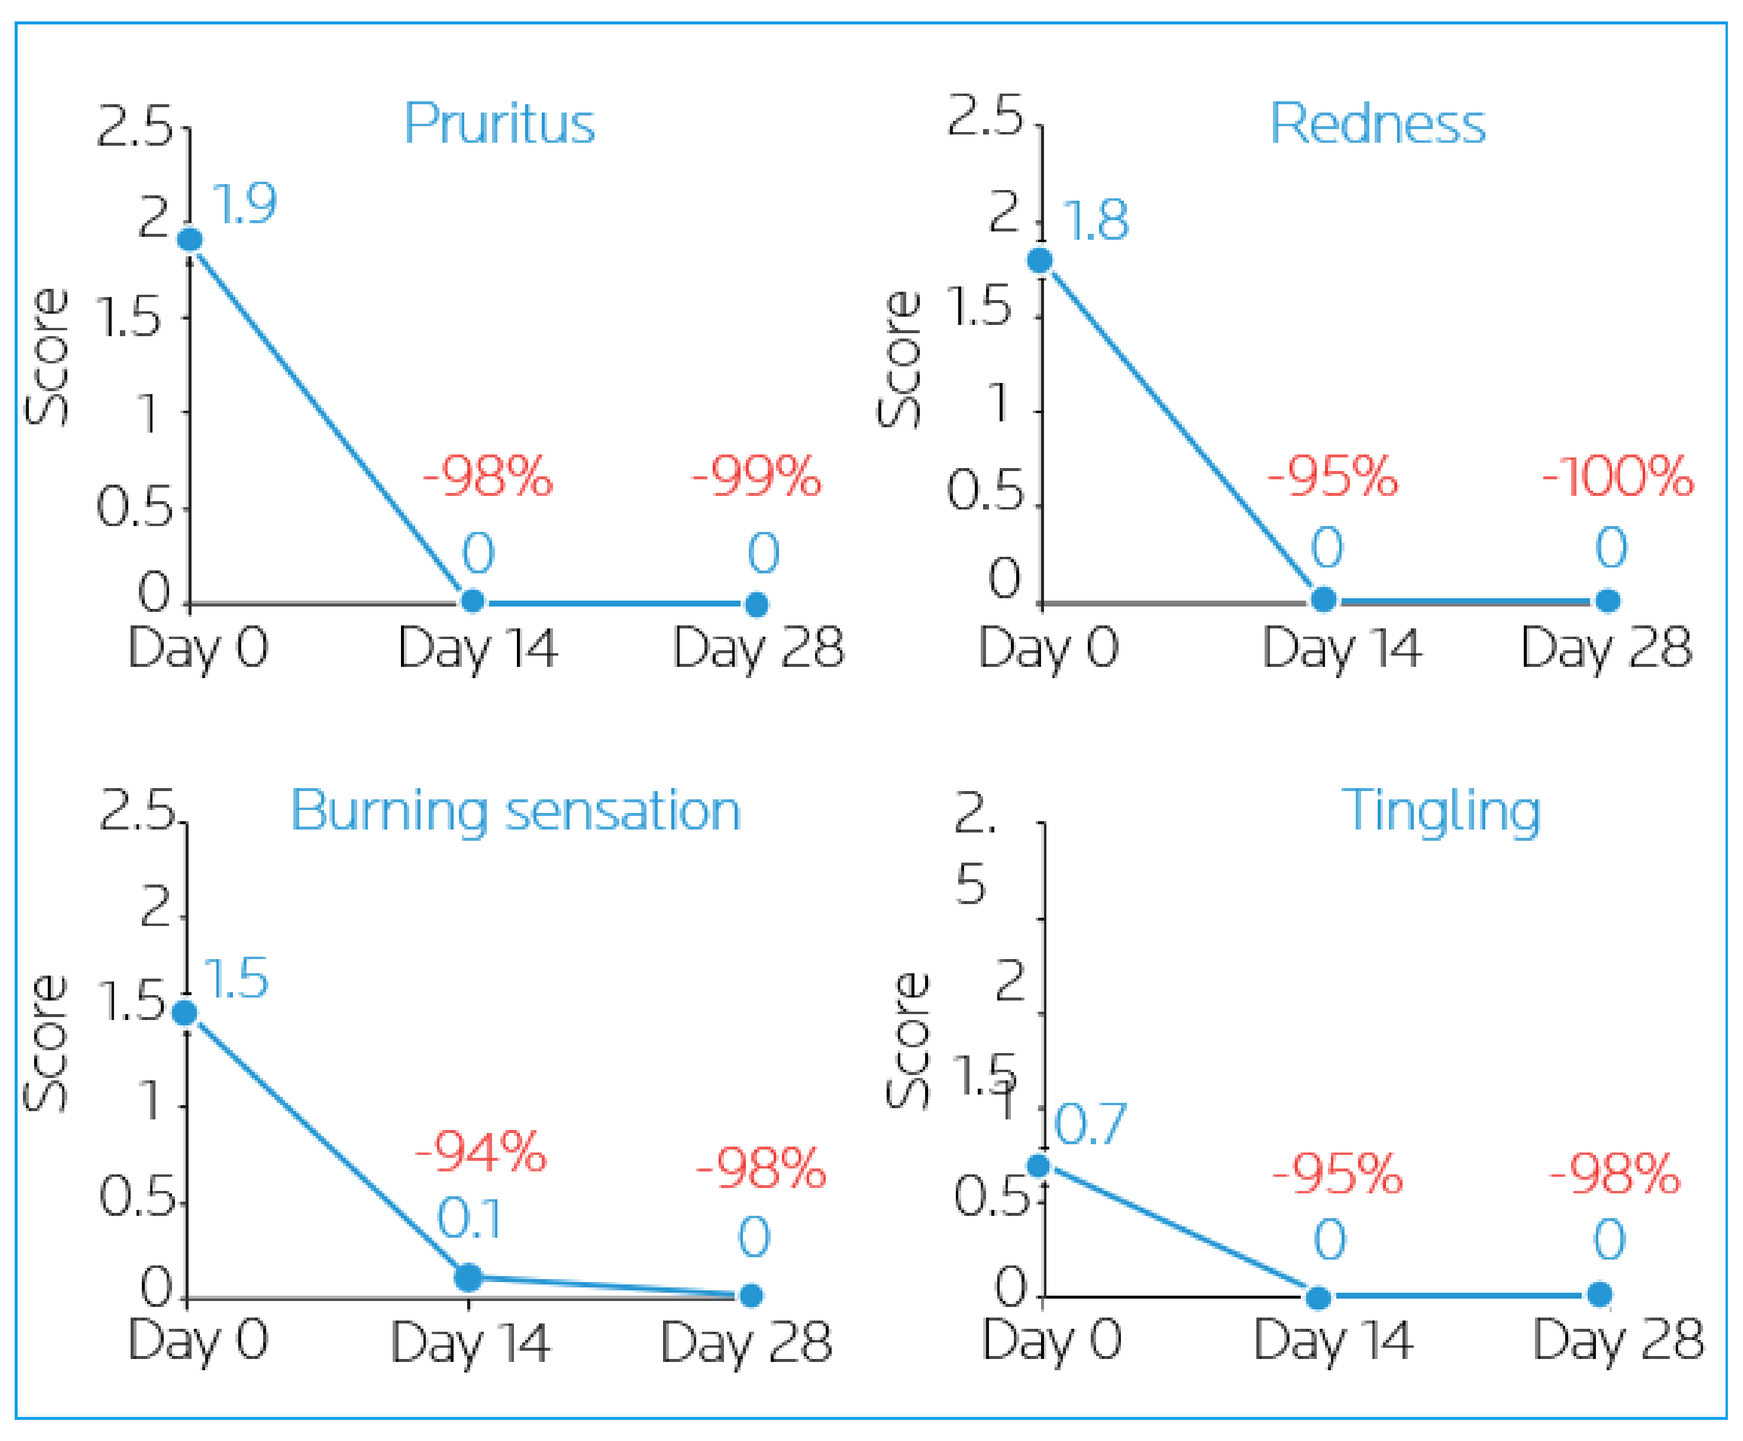
Pruritus
Score
1.9
-98%
0
-99%
0
Redness
1.8
-95%
0
-100%
0
Burning sensation
1.5
-94%
0.1
-98%
0
Tingling
0.7
-95%
0
-98%
0
Day 0
Day 14
Day 28

Subject-reported symptoms significantly (p<0.0001) improved as early as Day 14.

Figure 3

EVOLUTION OF TRANSEPIDERMAL WATER LOSS

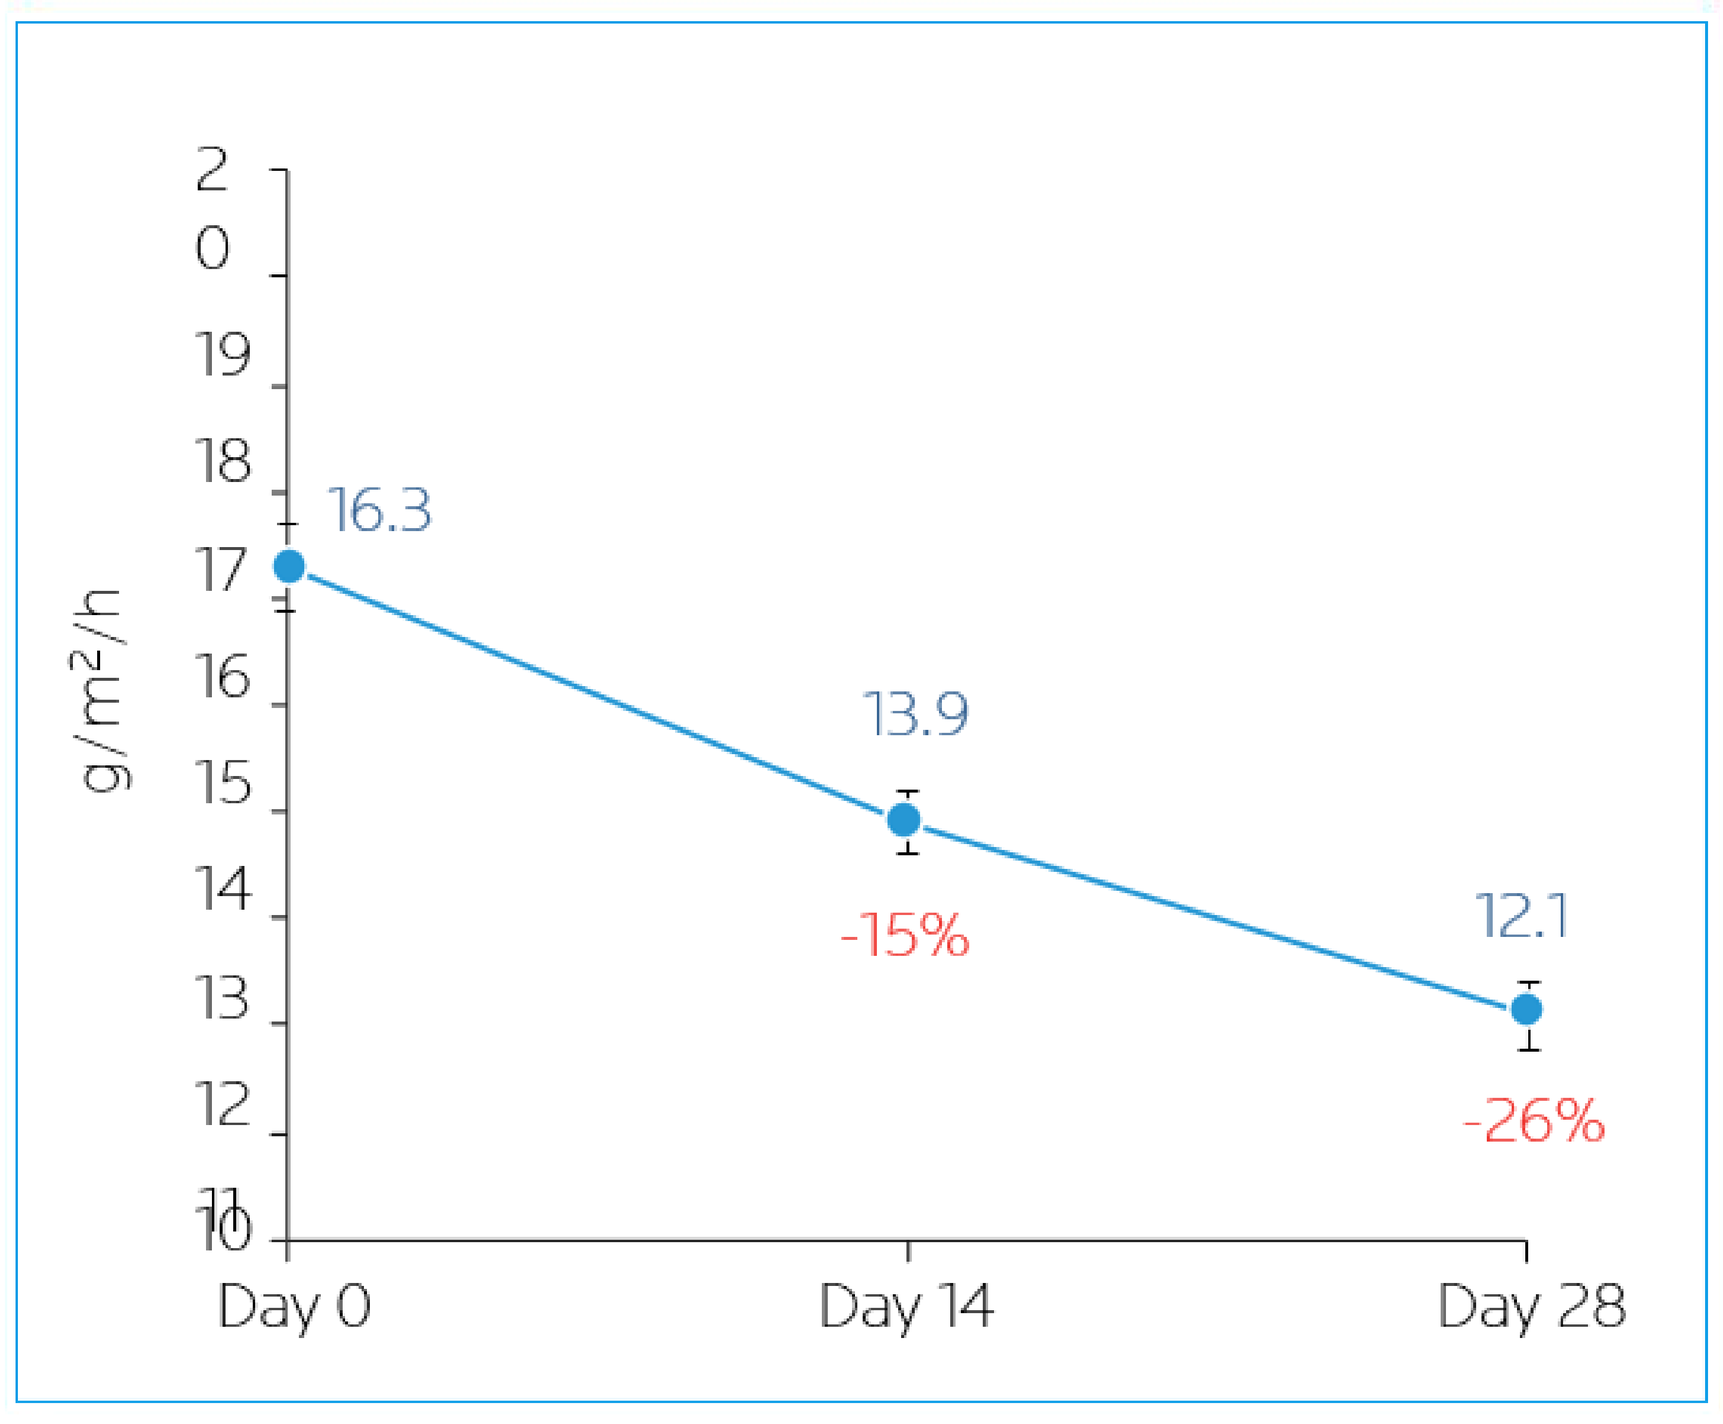
g/m²/h
16.3
13.9
-15%
12.1
-26%
Day 0
Day 14
Day 28

Transepidermal water loss was significantly (p<0.0001) reduced over time indicating a reinforcement of the cutaneous barrier.

Figure 4

EVOLUTION OF SKIN HYDRATION (CORNEOMETRY)

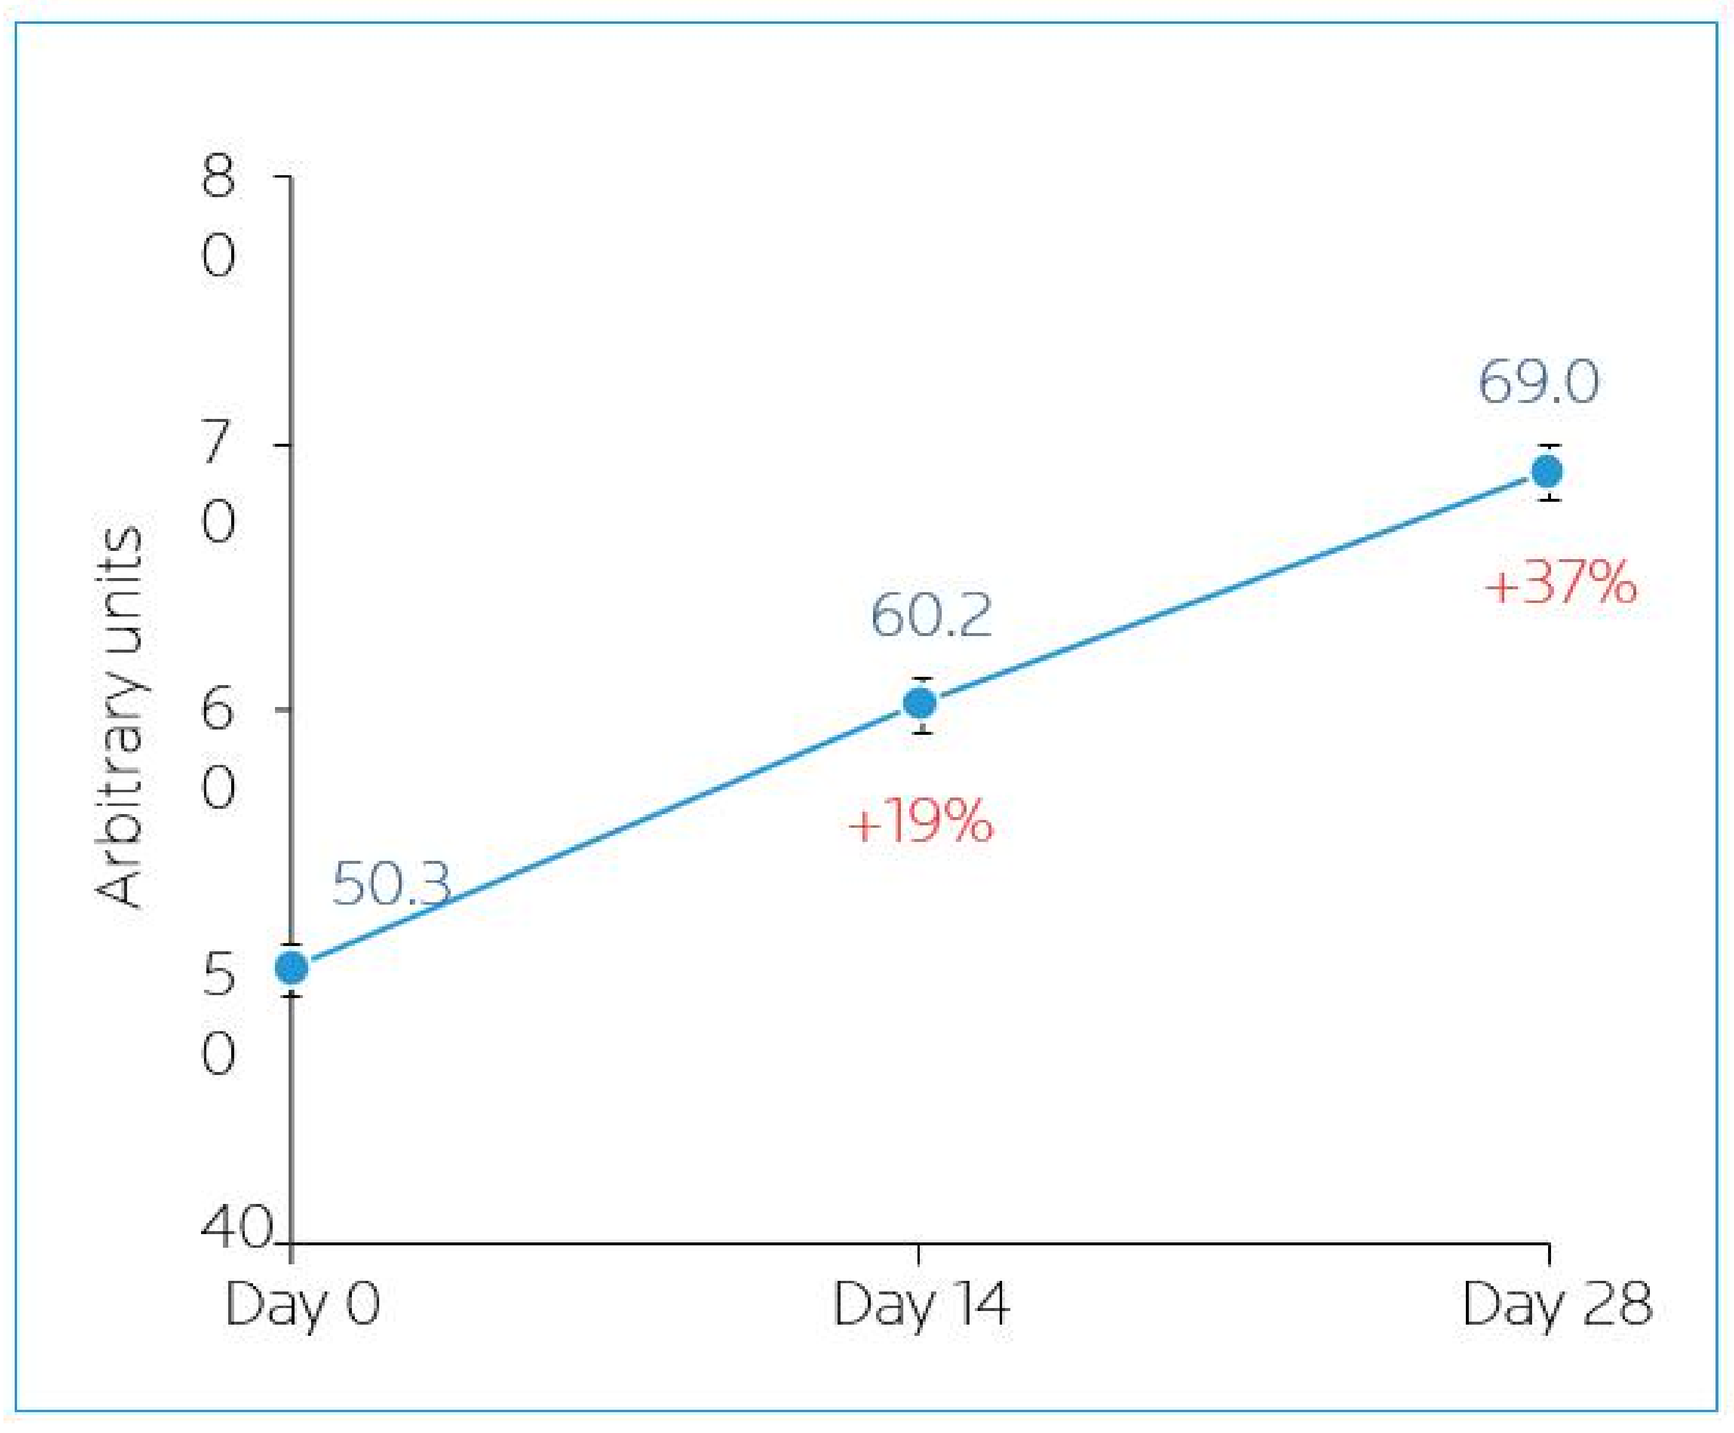
Arbitrary units
50.3
60.2
+19%
69.0
+37%
Day 0
Day 14
Day 28

Cutaneous hydration had significantly (p<0.0001) improved as early as Day 14.

DISCUSSION

A specifically formulated DC for intolerant/allergic skin is able to improve skin sensitivity signs and symptoms in parallel to improve instrumental assessment of skin barrier

Acknowledgements:

We thank patients and investigators who took part in this study

Funding:

25th World Congress of Dermatology SINGAPORE 2023

LA ROCHE-POSAY LABORATOIRE DERMATOLOGIQUE

PLATINUM SPONSOR

References:

1. Zirwas MJ. Contact Dermatitis to Cosmetics. Clin Rev Allergy Immunol. 2019;56(1):119-28.
2. Park ME, Zippin JH. Allergic contact dermatitis to cosmetics. Dermatol Clin. 2014;32(1):1-11.
3. Biebl KA, Warshaw EM. Allergic contact dermatitis to cosmetics. Dermatol Clin. 2006;24(2):215-32, vii.
4. Egawa G, Kabashima K. Barrier dysfunction in the skin allergy. Allergol Int. 2018;67(1):3-11.
5. Goleva E, Berdyshev E, Leung DY. Epithelial barrier repair and prevention of allergy. J Clin Invest. 2019;129(4):1463-74.
6. Di Agosta E, Salvati L, Corazza M, Baiardini I, Ambrogio F, Angileri L, et al. Quality of life in patients with allergic and immunologic skin diseases: in the eye of the beholder. Clin Mol Allergy. 2021;19(1):26.
7. Boo YC. Mechanistic Basis and Clinical Evidence for the Applications of Nicotinamide (Niacinamide) to Control Skin Aging and Pigmentation. Antioxidants (Basel). 2021;10(8).